SAFETY, PHARMACOKINETICS AND PHARMACODYNAMICS OF SNF472 IN HEMODIALYSIS PATIENTS: NEW DATA FROM A PHASE 1B/2A RANDOMISED PLACEBO-CONTROLLED CLINICAL TRIAL

C. Salcedo[1], J. Perelló[1,2], R. Ojeda[3], P.H. Joubert[1,4], M. Arias[3], AZ. Canals[1], M.D. Ferrer[1], M.M. Pérez[1], V. Torregrosa[3], JM. Campistol[3], F. Maduell[3]

[1]Laboratoris Sanifit SL, Palma, Spain; [2]University of the Balearic Islands, Palma, Spain; [3]Nephrology Department, Hospital Clinic, Barcelona, Spain; [4]Institute of Pharmaceutical Science. King's College, London, UK **Contact:** carolina.salcedo@sanifit.com

INTRODUCTION

SNF472 is an intravenous (i.v.) formulation of myo-inositol hexaphosphate, a molecule that inhibits calcification by binding to the growing sites of the hydroxyapatite (HAP) crystal. SNF472 is being developed for the prevention of vascular calcification in patients with end-stage renal failure on hemodialysis (HD). Non-clinical investigations and a single dose study in healthy volunteers and HD patients supported proceeding to a repeated dose trial in HD patients.

AIM

The aim of this double-blind, placebo-controlled, randomized multiple ascending dose clinical trial was to investigate the safety, tolerability, pharmacokinetics (PK) and pharmacodynamics (PD) of repeated i.v. doses of SNF472 in HD patients.

MATERIALS AND METHODS

Design: Sixteen stable HD patients were divided into two 8-subjects cohorts. In each cohort, the subjects were randomized to receive SNF472 (six subjects) or placebo (two subjects) intravenously. SNF472 was administered three times per week by a 4-hour infusion during the HD sessions. Study design is shown in Fig. 1.

Cohort 1: Eight subjects participated in a Multiple Ascending Dose (MAD), consisting of five Treatment Periods of one week each, with a 3-week washout period between doses. The administered SNF472 doses were: 1, 3, 5, 12.5 and 20 mg/kg.

Cohort 2: Eight subjects participated in a Multiple (Fixed Repeated) Dose Clinical Trial, consisting of one single Treatment Period of four weeks and received either placebo (two subjects) or 10 mg/kg SNF472 (six subjects).

Determinations: Standard safety parameters (adverse events, clinical laboratory and haematology, clinical chemistry, vital signs, all 12 lead ECG parameters including cardiac intervals, PR, QRS, QT, and QTc, and physical examination), serum bio element concentrations (only in cohort 2), PK, and calcification PD (potential for ex vivo formation of HAP crystals in plasma samples obtained at baseline and at the end of the 4-hour of infusion) were determined.

Figure 1. Study design

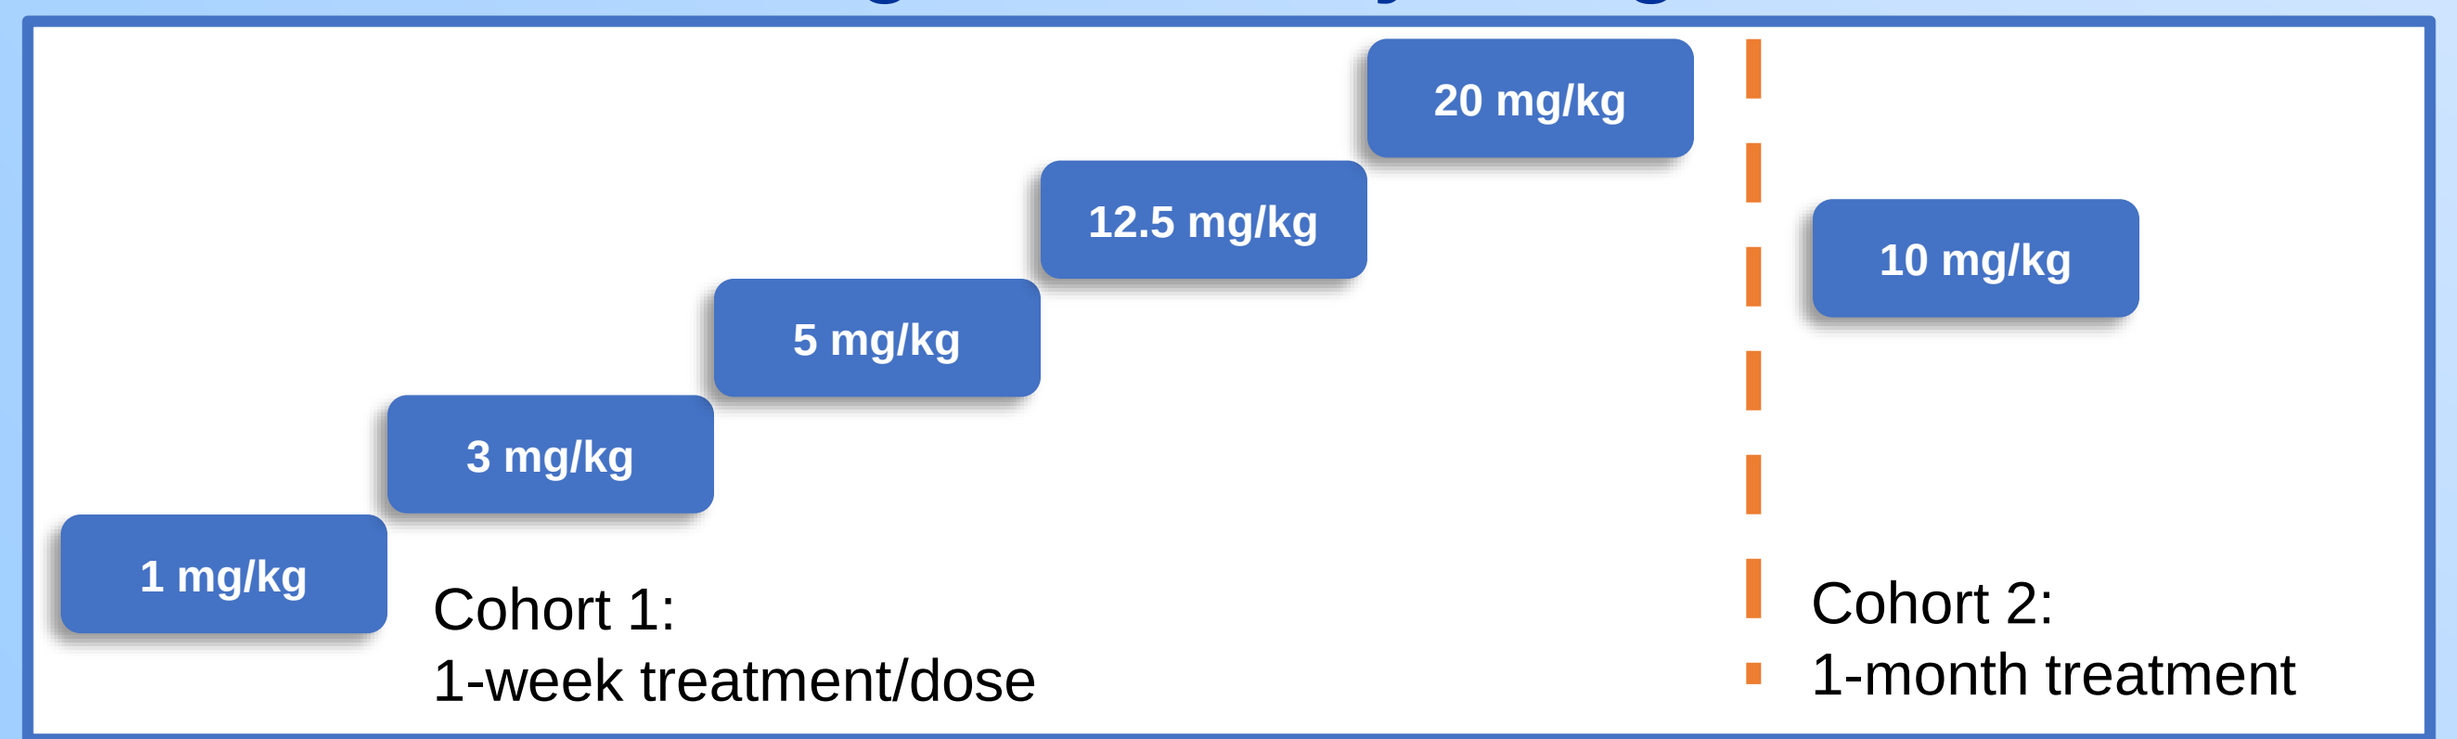
1 mg/kg
3 mg/kg
5 mg/kg
12.5 mg/kg
20 mg/kg
10 mg/kg
Cohort 1:
1-week treatment/dose
Cohort 2:
1-month treatment

Cofinanciado por el Fondo Europeo de Desarrollo Regional (FEDER). Unión Europea. Una manera de hacer Europa.
RTC-2014-2460-1

GOBIERNO DE ESPAÑA
MINISTERIO DE ECONOMÍA Y COMPETITIVIDAD

RESULTS

Pharmacokinetics: SNF472 PK is shown in Fig 2. C_{max} and AUC values increased with increasing doses of SNF472 after single and repeated administration in a slightly more than dose-proportional manner. There was no evidence of accumulation or metabolism induction after repeated administration for 26 days.

Figure 2. Pharmacokinetics of plasma SNF472 after 4-hour intravenous infusion in HD patients.

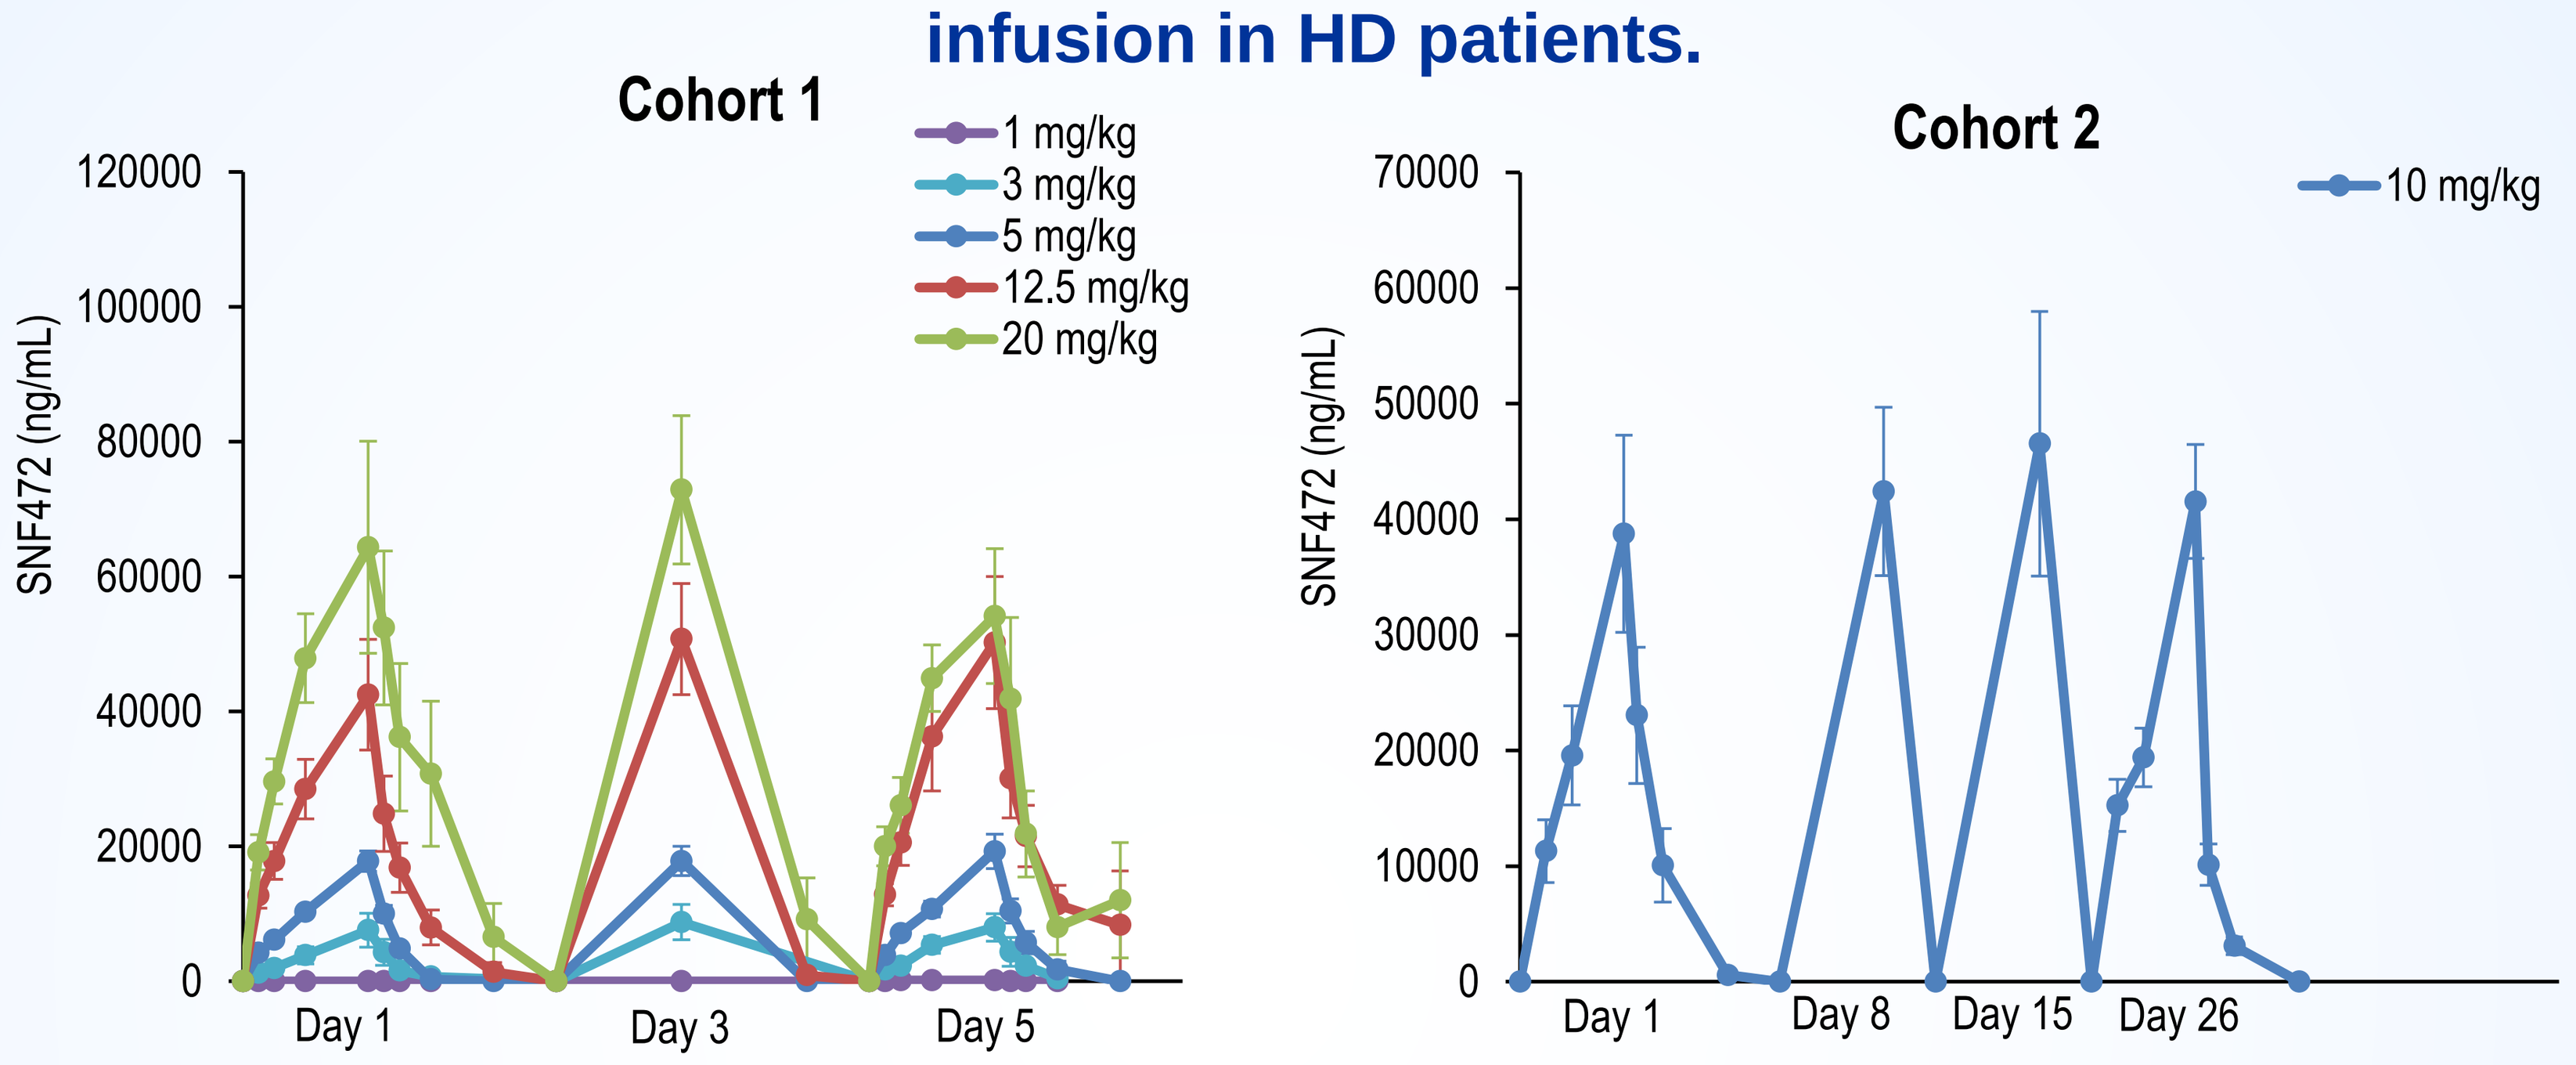
Cohort 1
1 mg/kg
3 mg/kg
5 mg/kg
12.5 mg/kg
20 mg/kg
SNF472 (ng/mL)
Day 1
Day 3
Day 5
Cohort 2
10 mg/kg
SNF472 (ng/mL)
Day 1
Day 8
Day 15
Day 26

Safety: No adverse events, systemic side effects or local irritation related to SNF472 administration were reported. Only one serious adverse event was reported but not related to treatment. No ionized calcium reduction was observed as a consequence of the treatment with SNF472 (Fig. 3). Therefore, SNF472 did not induce circulating calcium chelation. High variability in terms of QTcB was observed, but no test item correlation between ΔQTcB and C_{max} was detected. Serum P, Mg^{2+}, and K^{+} were decreased as a consequence of HD, and SNF472 did not affect these changes (Fig. 3).

Figure 3. Bio elements concentration in serum after SNF472 4-hour intravenous infusion in HD patients

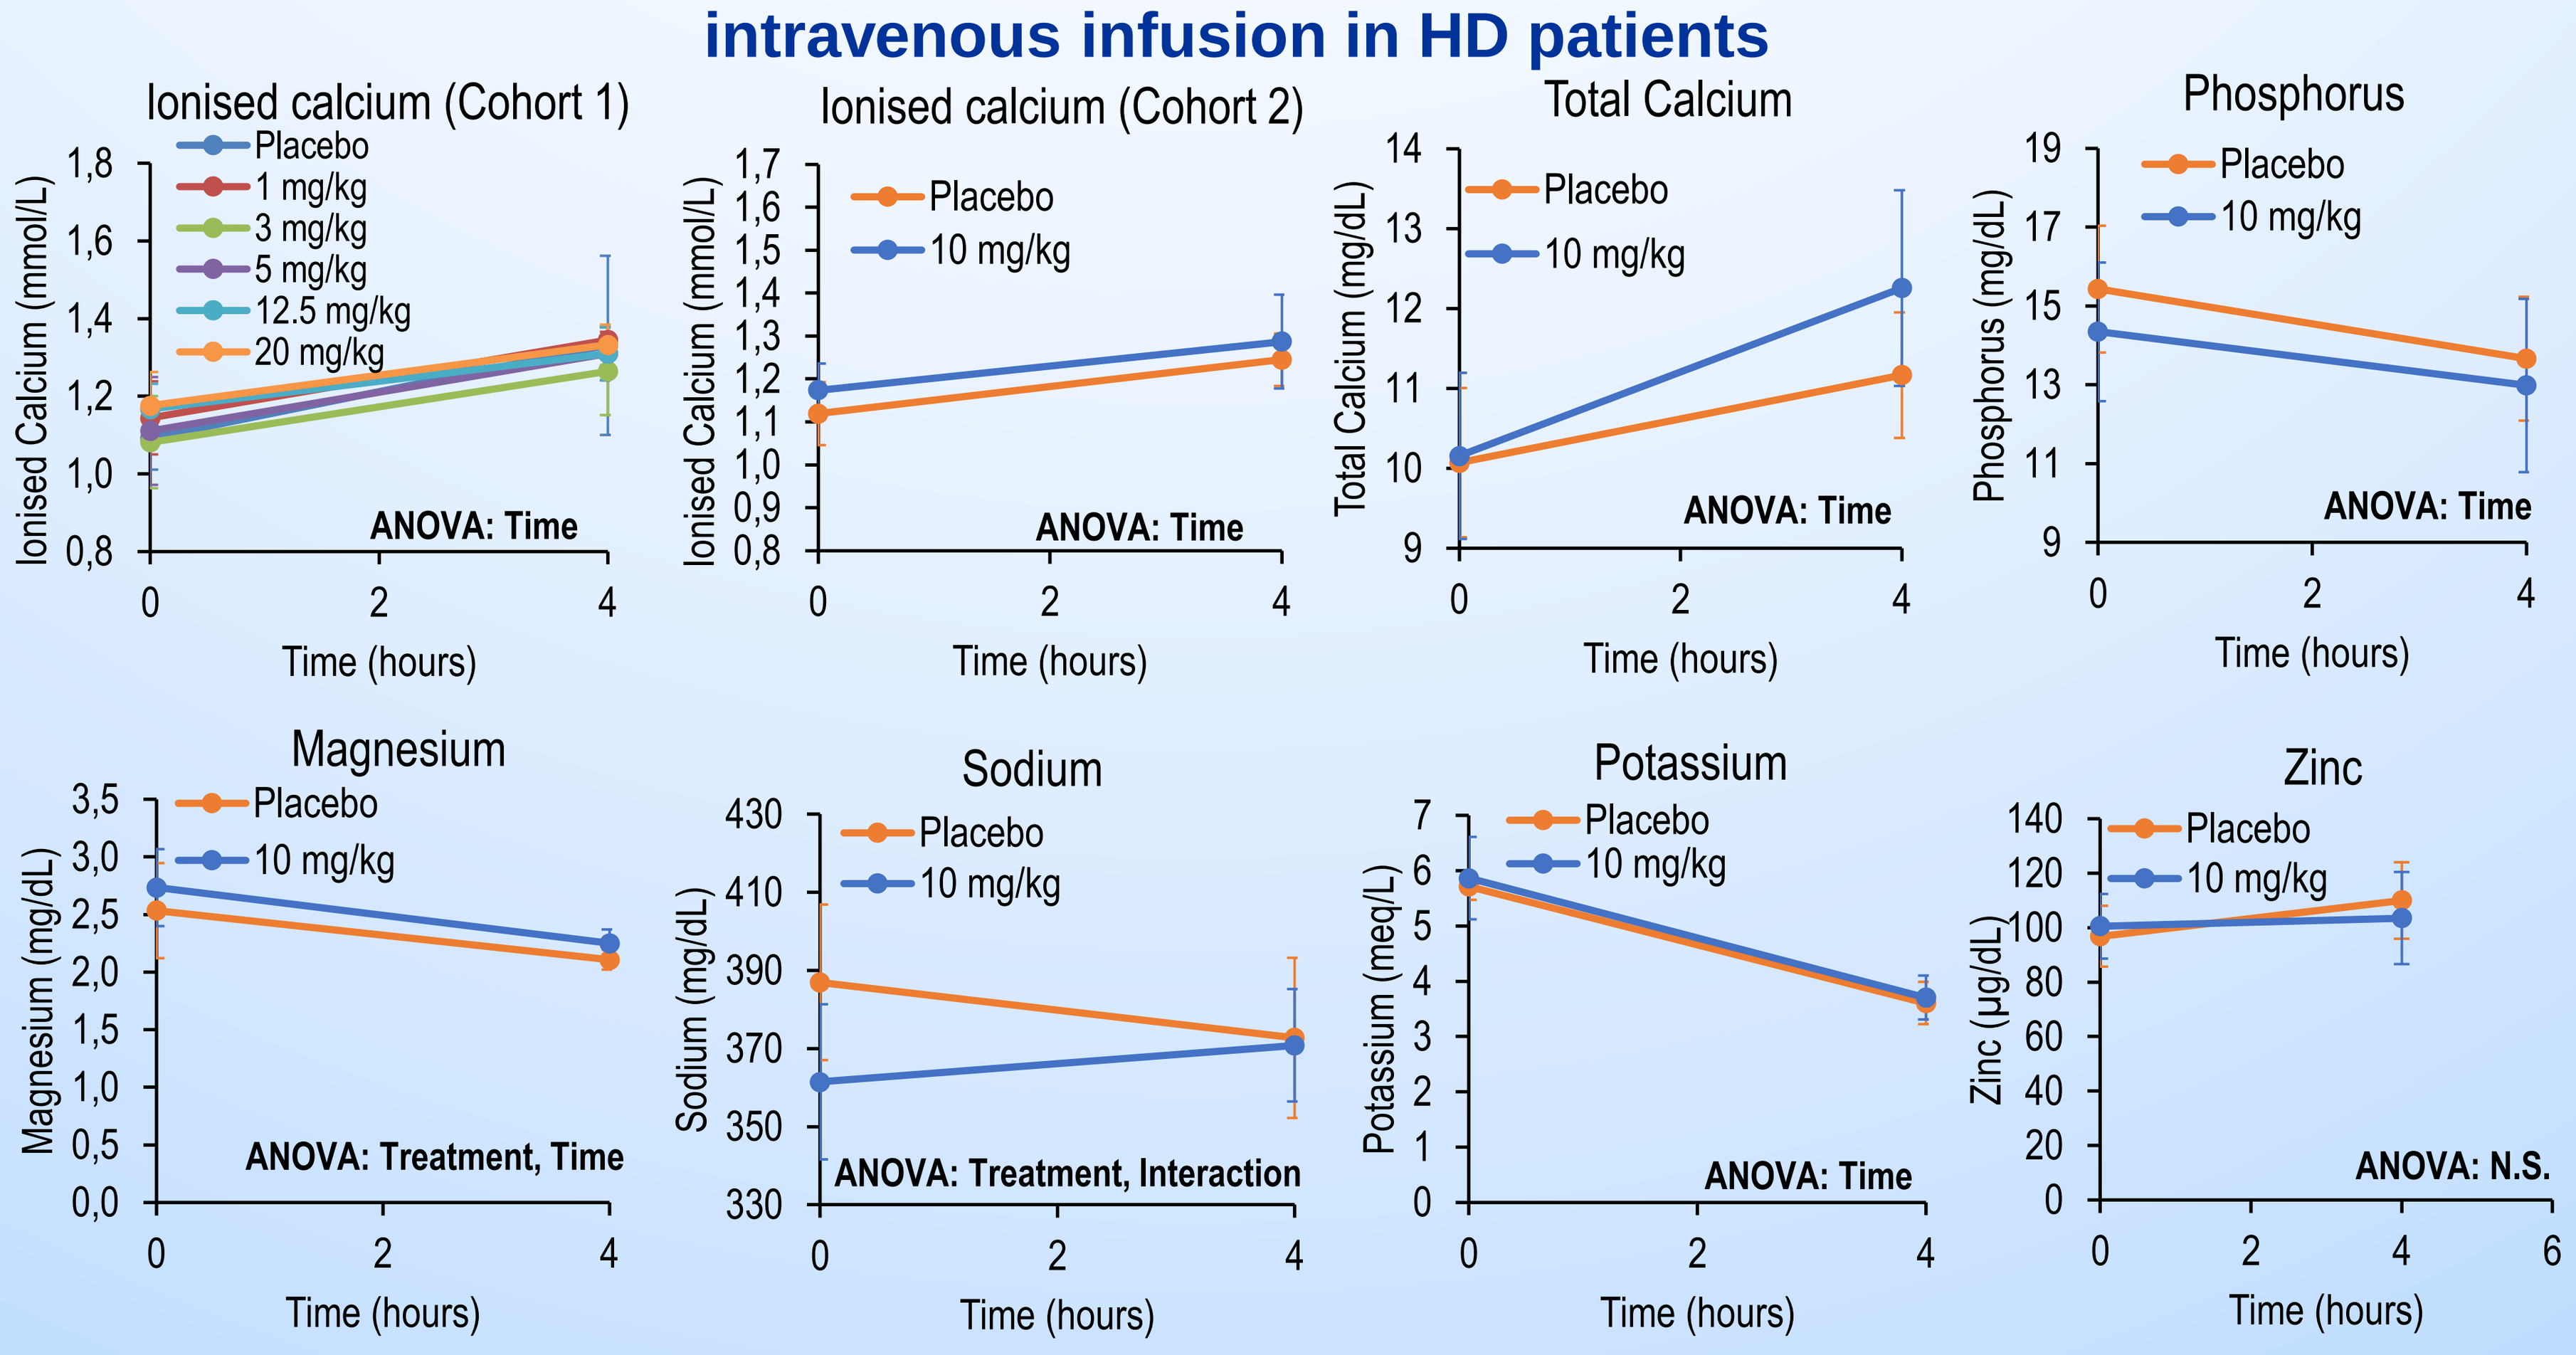
Ionised calcium (Cohort 1)
Placebo
1 mg/kg
3 mg/kg
5 mg/kg
12.5 mg/kg
20 mg/kg
ANOVA: Time
Ionised calcium (Cohort 2)
ANOVA: Time
Total Calcium
ANOVA: Time
Phosphorus
ANOVA: Time
Magnesium
ANOVA: Treatment, Time
Sodium
ANOVA: Treatment, Interaction
Potassium
ANOVA: Time
Zinc
ANOVA: N.S.
Placebo
10 mg/kg
Time (hours)

Statistical analysis: Two-way ANOVA. Factors: Time, Treatment. P < 0.05.

Pharmacodynamics: A significant effect of SNF472 on the inhibition of HAP crystallization was observed at doses of 3 mg/kg or higher (Fig. 4). No significant differences were observed when SNF472 was dosed between 3 and 20 mg/kg. The IC_{50} using this assay was 2.18 mg/kg.

Figure 4. Inhibition of induction of hydroxyapatite crystallization in plasma samples after SNF472 4-hour intravenous infusion in HD patients

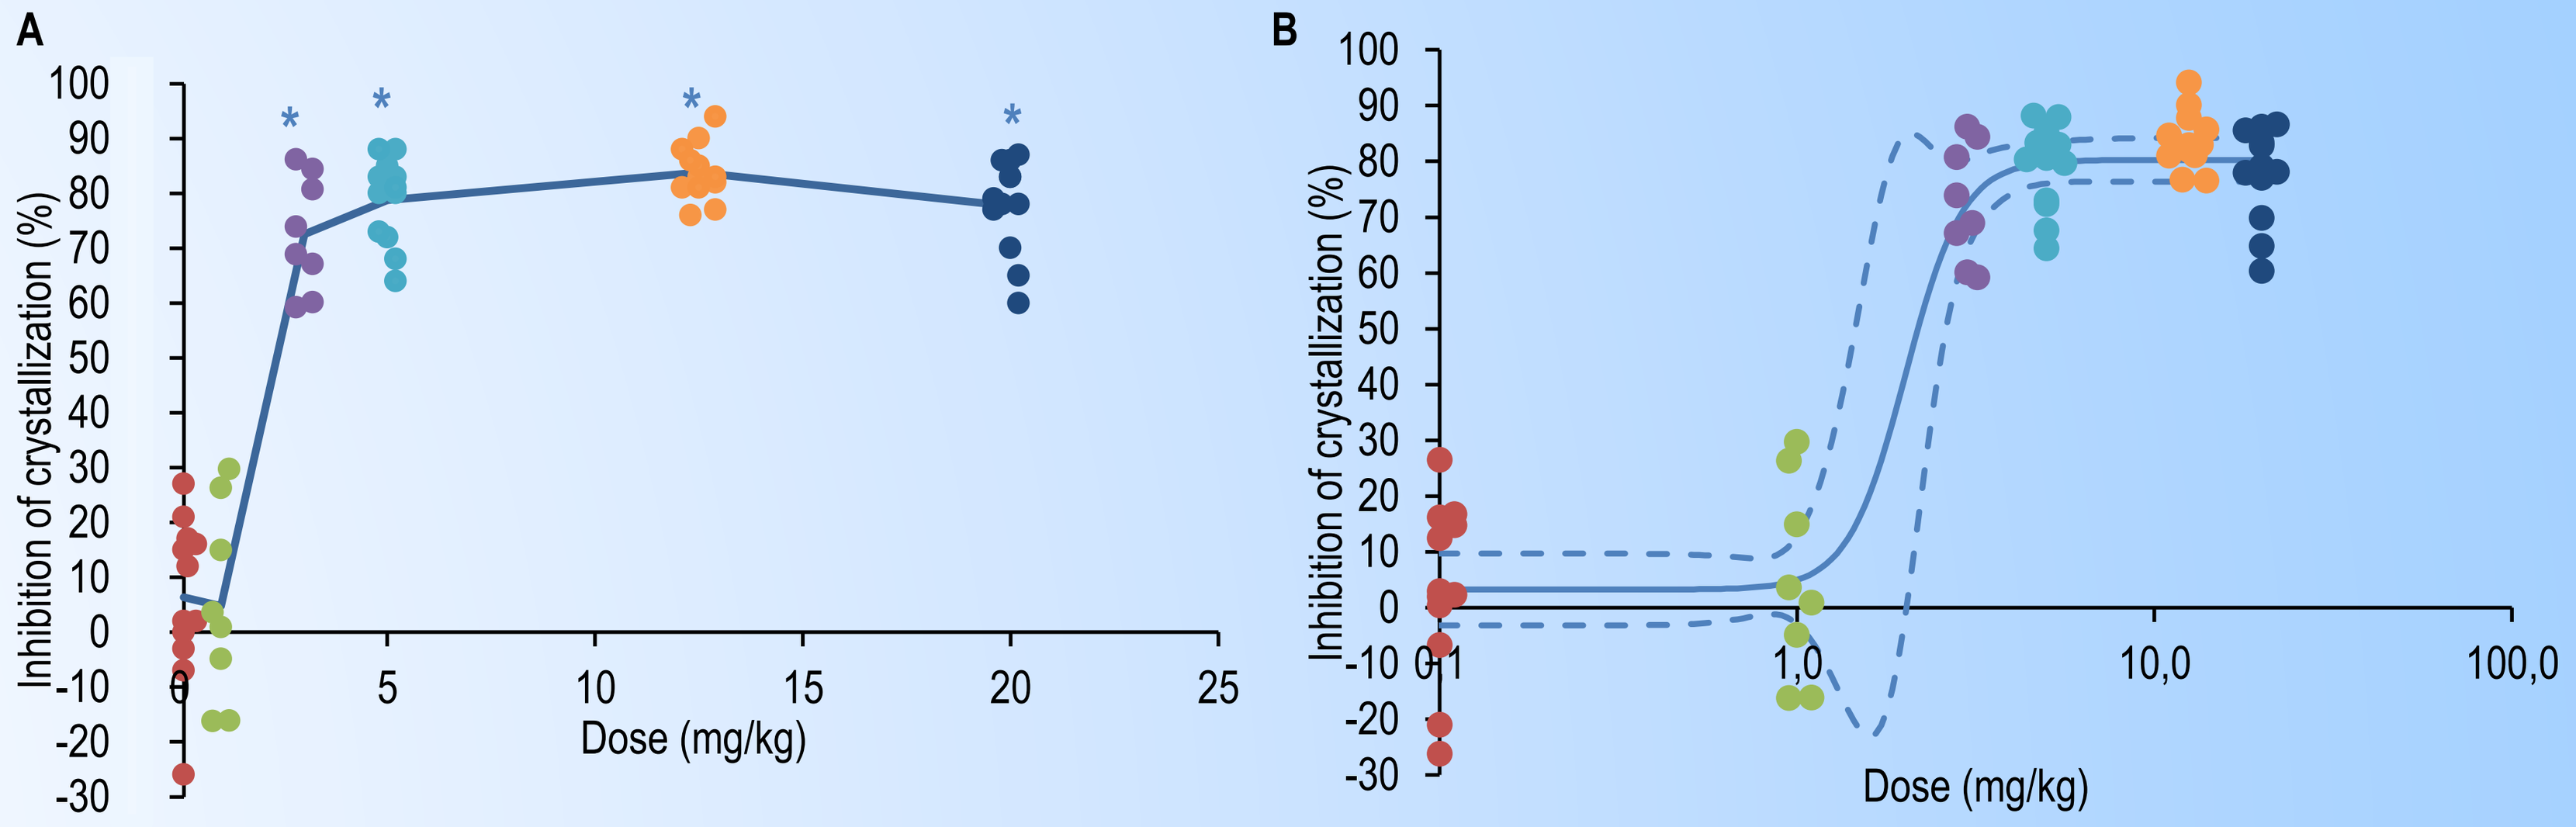
A
Inhibition of crystallization (%)
Dose (mg/kg)
B
Inhibition of crystallization (%)
Dose (mg/kg)

(A) Dose-response linear modelling. Solid line connects the means of the doses; (B) Four-parameter dose-response modelling. Solid line represent the dose-response curve calculated from the individual data points. Dashed lines represent the 90% confidence interval of the model. One-way ANOVA: (*) significant differences vs placebo and 1 mg/kg; p < 0.001.

A significant effect of SNF472 treatment (10 mg/kg vs placebo) evaluated on days 1, 8, 15 and 26 was also observed in cohort 2 patients (Fig. 5). Intravenous infusion of 10 mg/kg SNF472 inhibited HAP crystallization to a mean 78% throughout the 4 weeks of study.

Figure 5. Inhibition of induction of hydroxyapatite crystallization in plasma samples after SNF472 4-hour intravenous infusion in HD patients over 4 weeks

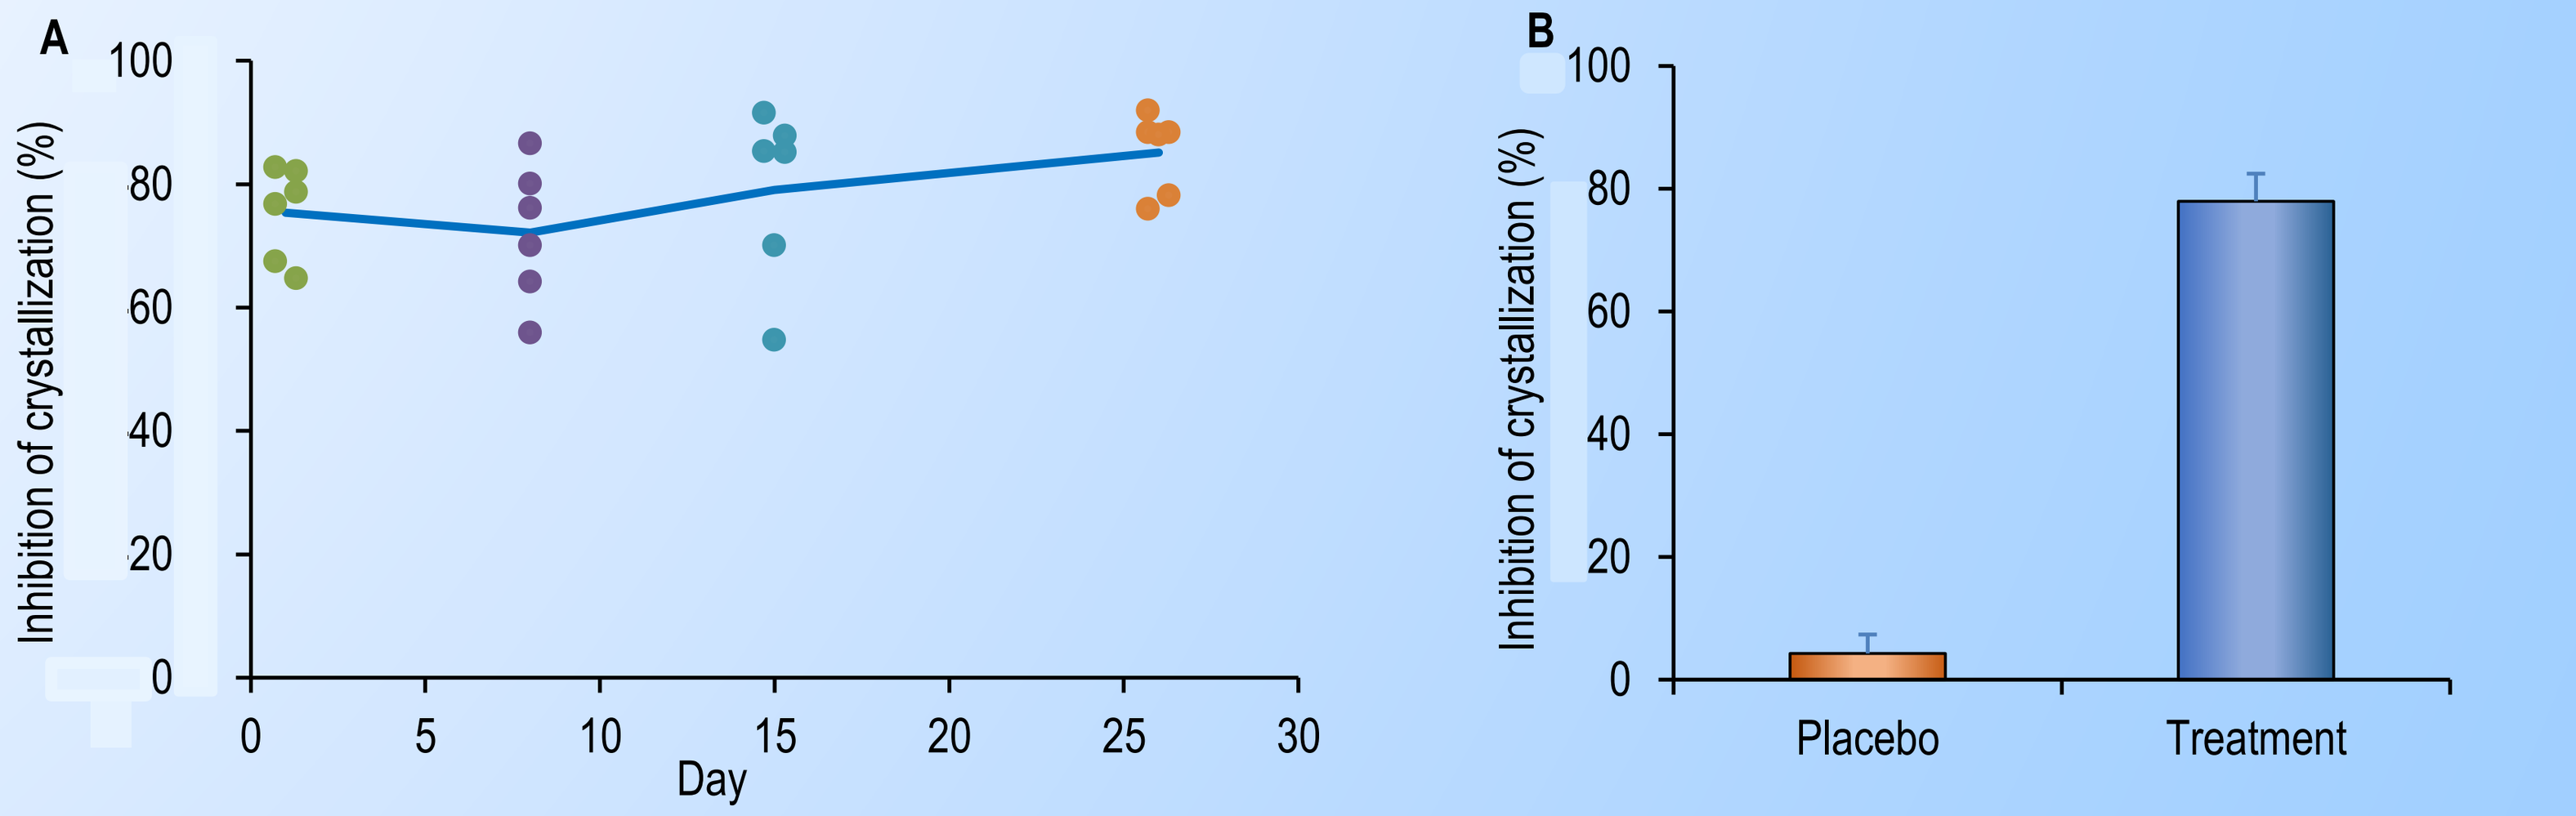
A
Inhibition of crystallization (%)
Day
B
Inhibition of crystallization (%)
Placebo
Treatment

(A) Time-course of inhibition in active patients over the 4-week period. Solid line connects the means. (B) Global inhibition in the whole study period. Results represent mean ± S.E.M. Student's T-test: (*) significant differences vs. placebo, p ≤ 0.001.

CONCLUSIONS

1. **SNF472 showed an adequate PK profile, suggesting low clearance through the dialysis membrane**
2. **SNF472 showed good safety at all tested doses, up to 20 mg/kg (C_{max} 70,000 ng/mL / 105 µM)**
3. **SNF472 reduced plasma calcification propensity in HD patients with an IC_{50} of 2.18 mg/kg**