SNF472 INHIBITS CARDIOVASCULAR CALCIFICATION IN UREMIC RATS

J. Perelló[1,2], C. Salcedo[1], E. Neven[3], G.J. Behets[3], P.H. Joubert[1], P.C. D'Haese[3], M.D. Ferrer[1]

[1] Laboratoris Sanifit SL. Research and Development Department, 07121 Palma de Mallorca, Spain
[2] Laboratory of Renal Lithiasis Research, IUNICS, University of the Balearic Islands, 07122 Palma de Mallorca, Spain
[3] Laboratory of Pathophysiology, University of Antwerp, Belgium
Contact: joan.perello@sanifit.com

KIDNEYWEEK 2015 San Diego, CA • Nov 3-8

INTRODUCTION

SNF472 is an intravenous (i.v.) formulation of myo-inositol hexaphosphate, a small and highly water-soluble molecule that inhibits calcification by binding to the growing sites of the hydroxyapatite (HAP) crystal. SNF472 has been shown to inhibit the development and progression of cardiovascular calcification (CVC) in non-uremic rats[1,2]. SNF472 is in active clinical development.

AIM

To investigate the pharmacokinetics (PK) and efficacy (inhibition of CVC) of i.v. SNF472 in control and uremic rats.

MATERIALS AND METHODS

Pharmacokinetics study (PK)

The study was performed with 10 male Wistar rats in which uremia was induced by 10 daily oral (p.o.) administrations of 600 mg/kg of adenine (days 1 – 10) and oral administration of 300 ng/kg α-calcidol on days 11 and 13, following the model described by Terai[3]. Animals were previously distributed at random into 2 groups, which received 10 or 50 mg/kg of SNF472 daily as a 4-hour intravenous infusion. Blood was collected at days 1 (controls) and 14 (uremic) at similar time points during the 4-hour i.v. infusion and SNF472 plasma concentrations were determined by UPLC®-MS analysis.

Efficacy study (EF)

Uremia was induced to 24 male Wistar rats as above whilst α-calcidol was administered 3 times/week from day 11 to day 19. A total of 12 control rats received daily 4-hour i.v. infusions of 50 mg/kg SNF472 while 12 rats received saline from day 0 to day 19. At day 19 all surviving animals were sacrificed and blood and tissue samples collected to measure SNF472 plasma levels at C_{max} and calcium content in aorta and heart by ICP-OES after acid digestion.

RESULTS

***PK*:** At 10 mg/kg, steady state (SS) was not reached in control animals (CA) during SNF472 4 hours infusion and maximum concentration (C_{max}) was found at the end of infusion. In contrast to this, maximum plasma concentration was reached at 30 minutes in uremic animals (UA). A low plasma level SS was reached at 60 min with a 7 times lower exposure than CA (Figure 1A and Table 1).

Figure 1. Pharmacokinetics of SNF472 after intravenous infusion in control and adenine-induced uremic rats

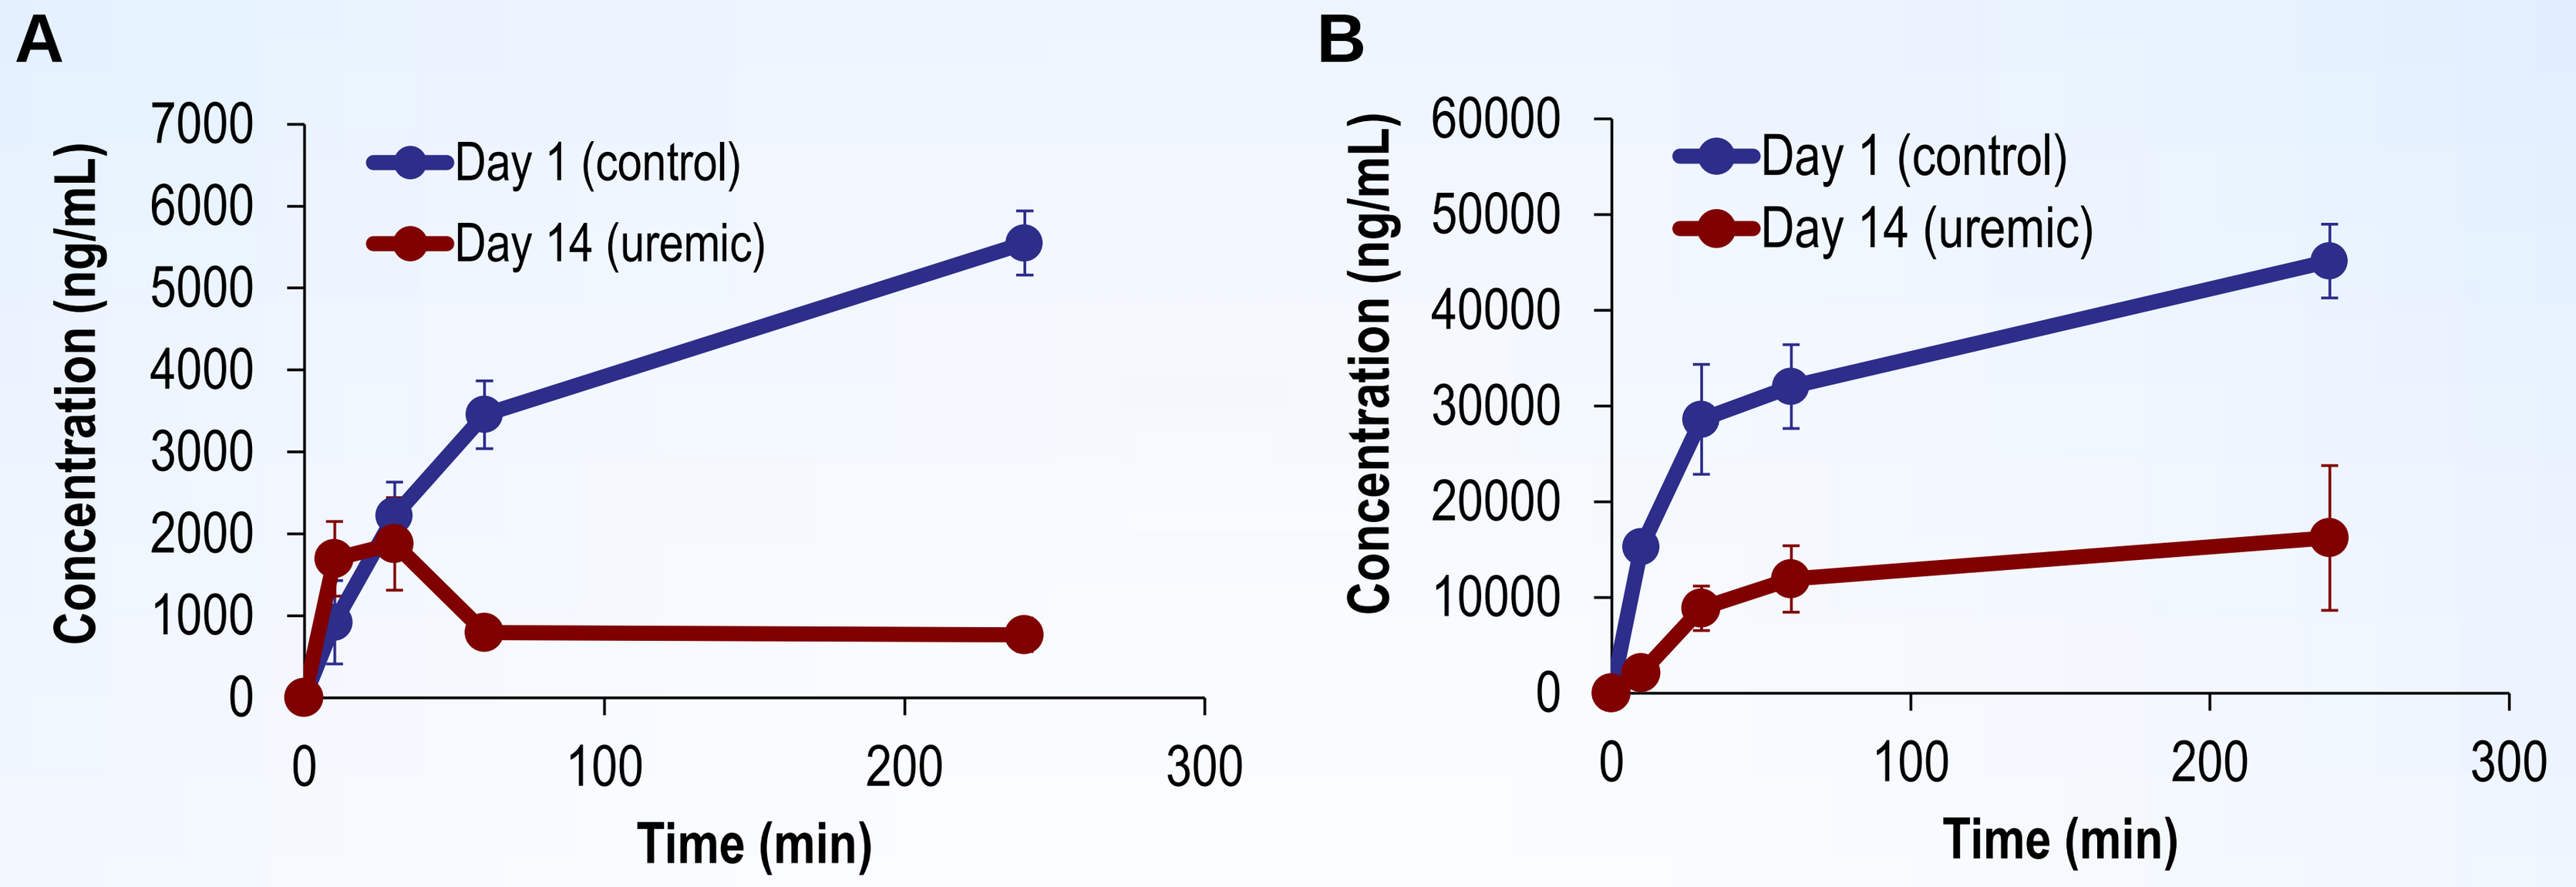

A
B
Concentration (ng/mL)
Day 1 (control)
Day 14 (uremic)
Time (min)

(A) Administration of 10 mg/kg SNF472; (B) administration of 50 mg/kg SNF472. Mean ± SEM.

Table 1. Pharmacokinetic parameters after SNF472 intravenous infusion administration in control and adenine-induced uremic rats

	Day 1 (control)		Day 13 (uremic)	
	10 mg/kg	50 mg/kg	10 mg/kg	50 mg/kg
C_{max} (ng/mL)	5548	45141	1877	16218
$AUC_{(0\to t)}$ (ng·h/mL)	16027	145212	3877	51665

C_{max}: maximum concentration measured. AUC: area under the curve

At 50 mg/kg SS was reached at 60 min in CA and at 30 minutes in UA. 3 times lower exposure than CA was seen in UA, as observed for 10 mg/kg (Figure 1B,Table 1). The levels reached at 50 mg/kg in UA animals were still over the expected therapeutic range (5,000-10,000 ng/mL) confirmed in previous preclinical studies and in Phase 1 clinical trials by means of pharmacodynamic measurements.

***EF*:** All animals developed uremia as a consequence of the adenine dosing in all treatment groups, as evidenced by the high plasma levels of urea and creatinine. A significant reduction (17%) in serum calcium levels was observed in the animals treated with the compound, while no changes in circulating phosphorus were evidenced (Figure 2).

Figure 2. Biochemistry parameters and SNF472 plasma levels in adenine-induced uremic rats

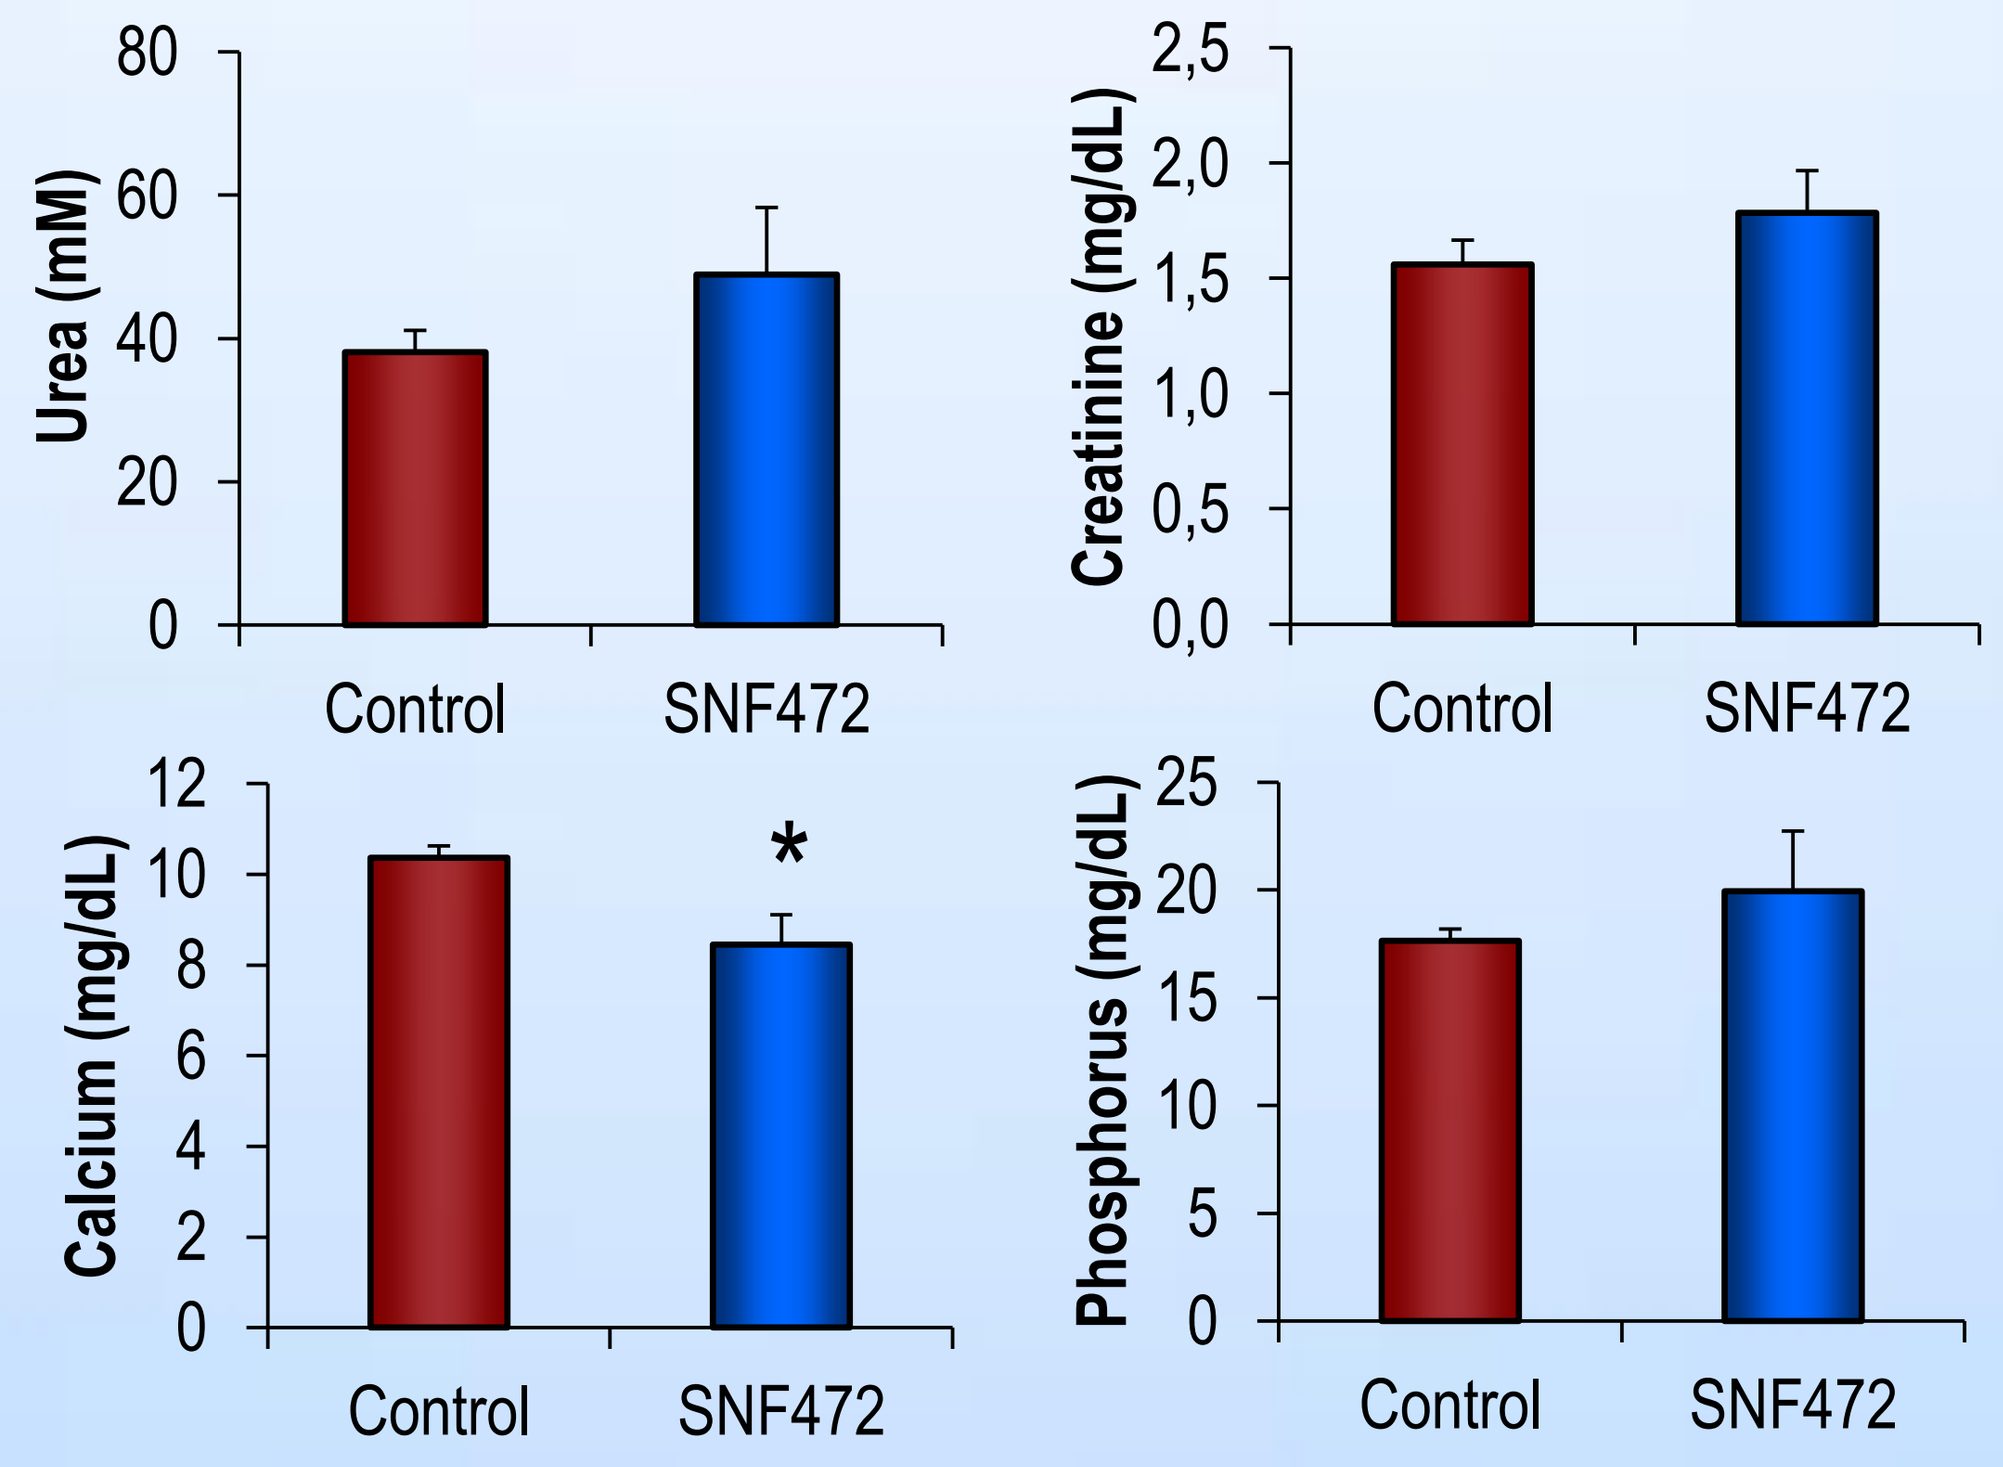

Urea (mM)
Creatinine (mg/dL)
Calcium (mg/dL)
Phosphorus (mg/dL)
Control
SNF472
*

Mean ± SEM. Statistical analysis: One-way ANOVA. (*) Differences vs control, $p < 0.05$

Taking into account the previous PK data, the dose was adjusted to obtain the plasma levels expected to reach efficacy, so SNF472 was tested at 50 mg/kg. Circulating SNF472 levels were measured prior and after adenine treatment and the decreased exposure in UA was confirmed, but still with high C_{max} around 20,000 ng/mL.

Aorta calcification was inhibited up to 80% in rats treated with SNF472 (Figure 3). Similar results were obtained concerning heart calcification, with an inhibition of 85%.

Figure 3. Aorta and heart calcium levels and inhibition of calcification in adenine-induced uremic rats treated with 50 mg/kg i.v. SNF472

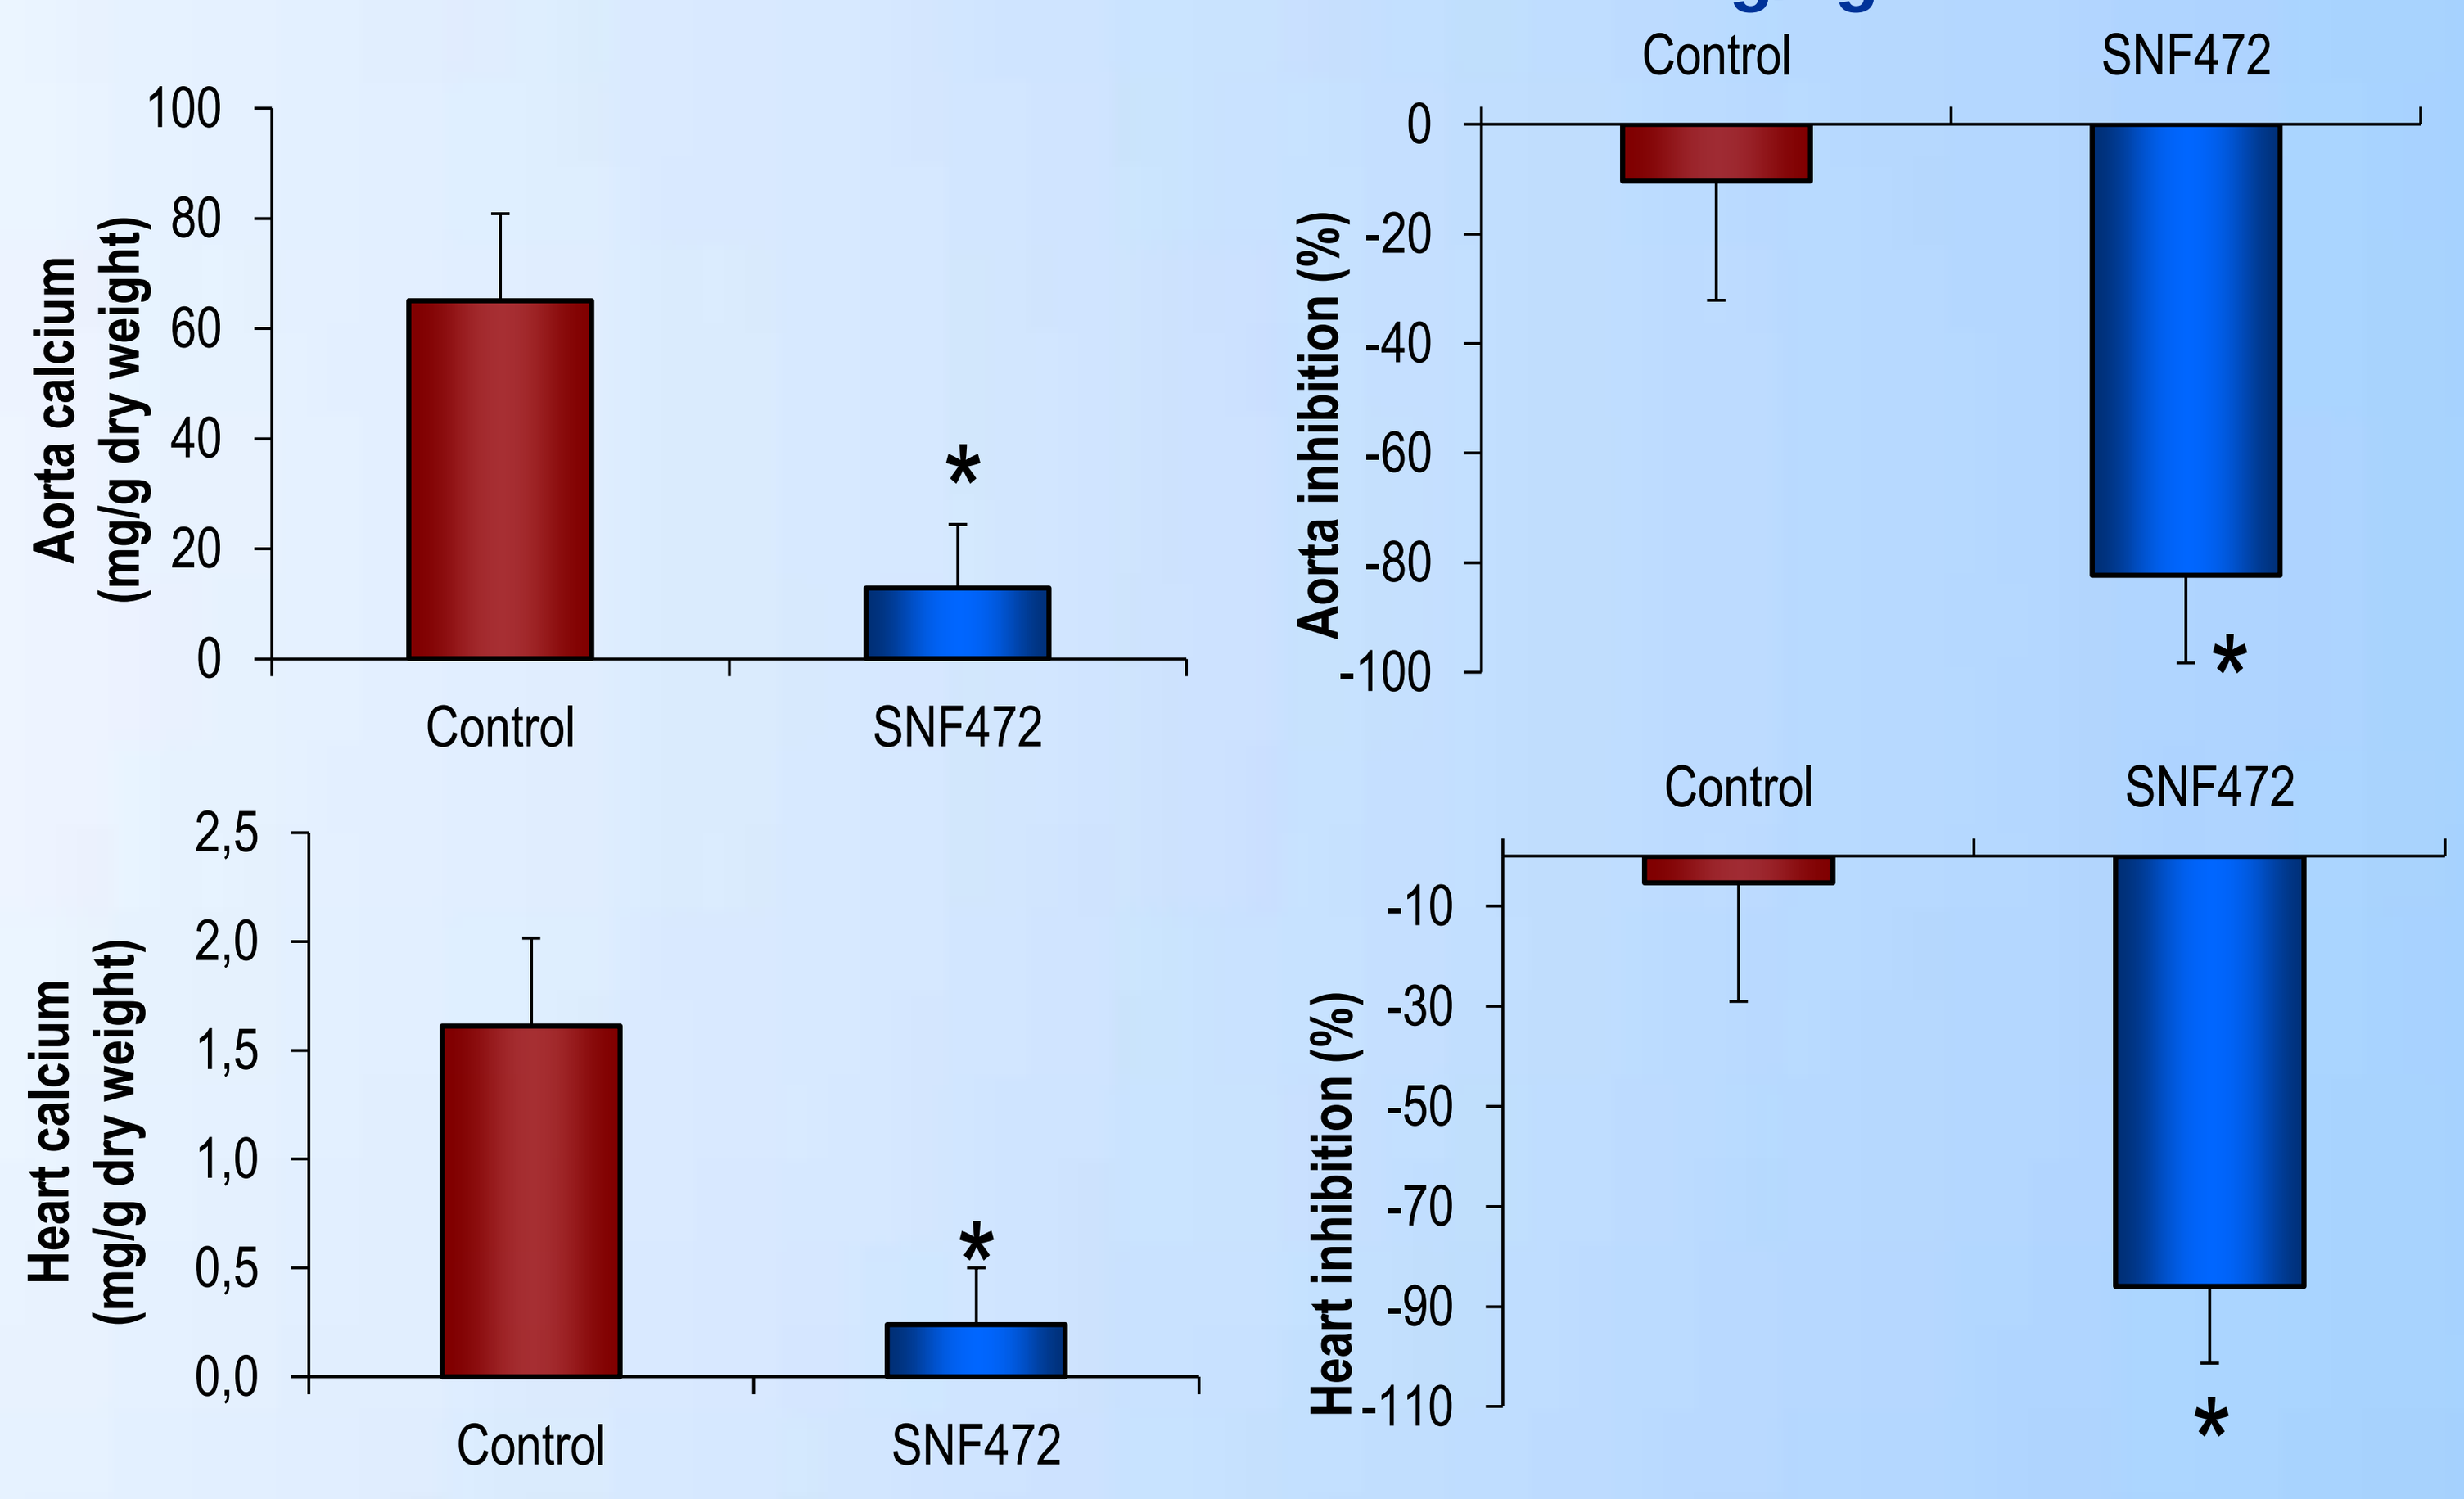

Aorta calcium (mg/g dry weight)
Aorta inhibition (%)
Heart calcium (mg/g dry weight)
Heart inhibition (%)
Control
SNF472
*

Median ± SEM. Statistical analysis: One-way ANOVA. (*) Differences vs control, $p < 0.05$

CONCLUSIONS

Daily 4 hour infusions of SNF472 inhibit the development of CVC by up to 85% in an adenine model of uremia in rats.

The exposure to SNF472 is reduced in these uremic rats but the therapeutic levels can be reached by adjusting the treatment dose.

These results support further investigation of SNF472 in the treatment of CVC in patients with calcification-related disorders such as calciphylaxis and ESRD patients.

BIBLIOGRAPHY

(1) J Am Soc Nephrol 2014; 25 (Abstract Supplement): 233A (**2**) Nephrol Dial Transplant 2015; 30 (suppl 3): iii588 **(3)** Br J Pharmacol 2009; 156: 1267-1278.