SNF472 IS A MOLECULE WITH THE POTENTIAL TO INHIBIT CARDIOVASCULAR CALCIFICATION IN END-STAGE RENAL DISEASE PATIENTS ON HEMODIALYSIS

Carolina Salcedo, Miguel D. Ferrer, Ana Z. Canals, Maria M. Pérez, Pieter H. Joubert, Joan Perelló

Laboratoris Sanifit SL. Research and Development Department, 07121 Palma de Mallorca, Spain – Contact: carolina.salcedo@sanifit.com

INTRODUCTION

SNF472 is an intravenous (i.v.) formulation of myo-inositol hexaphosphate (Fig. 1), a small and highly water-soluble molecule that inhibits calcification by binding to the growing sites of the hydroxyapatite (HAP) crystal, the main component of calcification in arteries. SNF472 is being developed for preventing **cardiovascular calcification** in end-stage renal disease (ESRD) patients undergoing **hemodialysis** (HD).

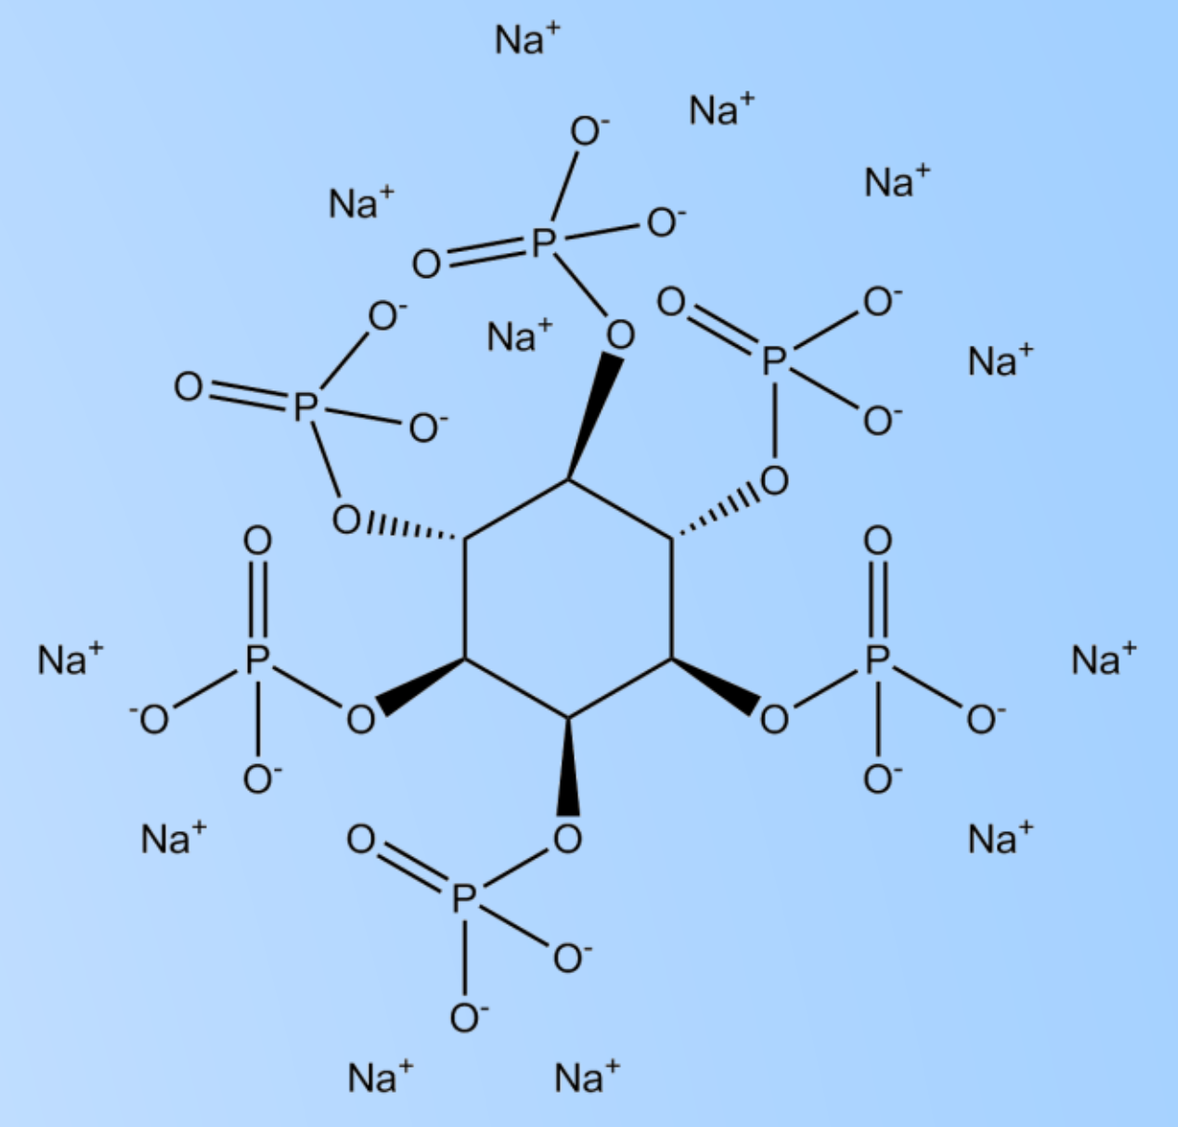

Figure 1. Chemical structure of SNF472

AIM

To assess the potential of SNF472 to inhibit cardiovascular calcification (CVC) in vivo in an animal model and ex vivo in HAP crystallization on HD patient samples.

PRECLINICAL EFFICACY STUDY

- Performed in 50 Sprague-Dawley rats with CVC induced by 75,000 IU/kg vitamin D.
- Five groups of 10 animals receiving: 0, 3, 10, 30, 100 mg/kg SNF472

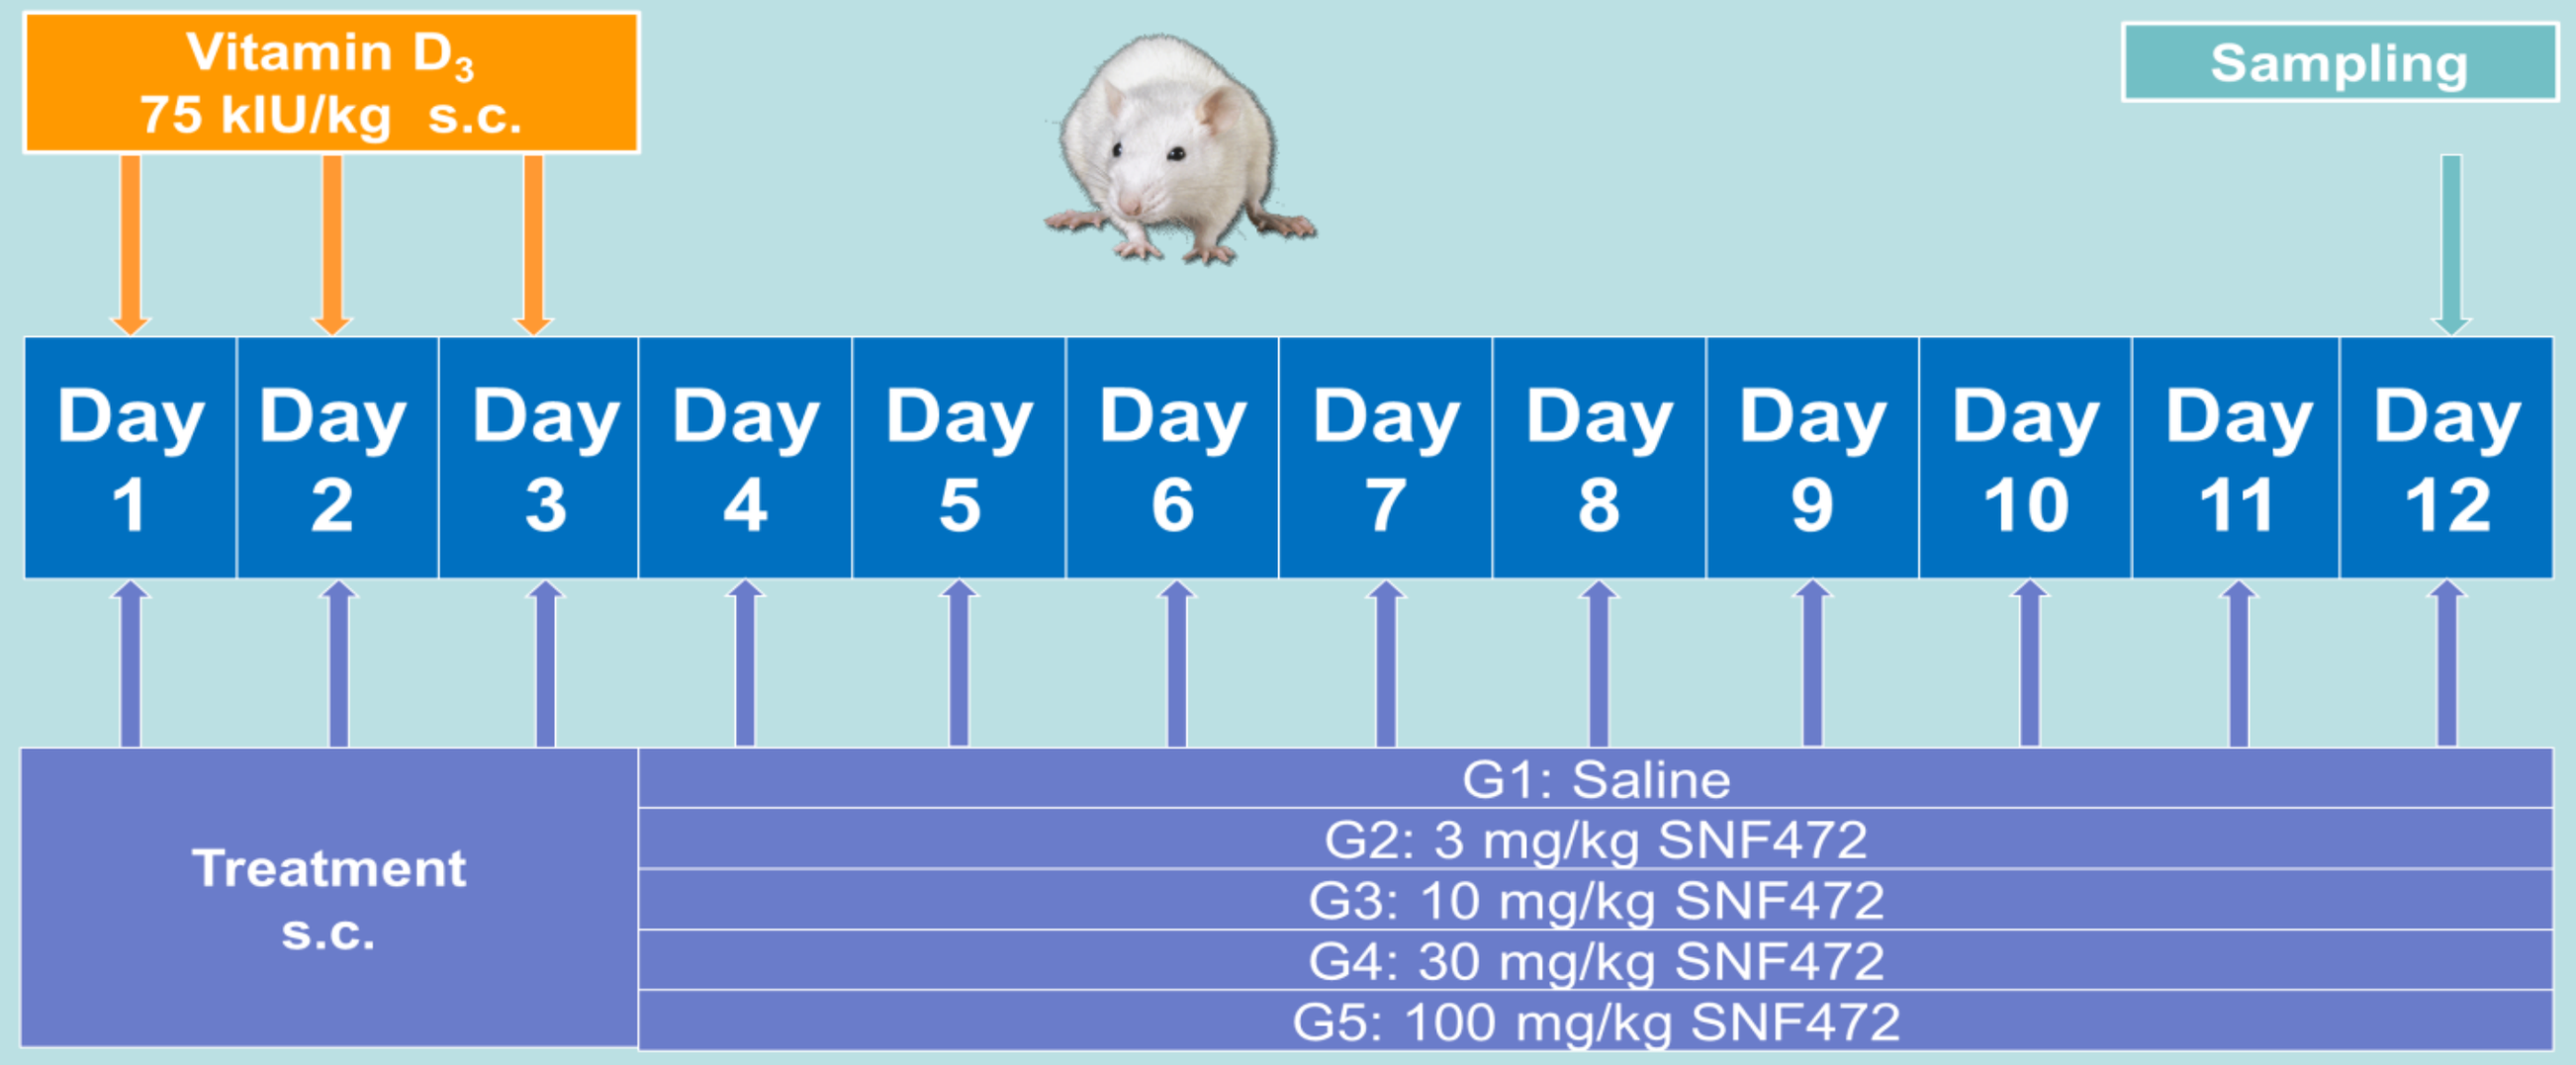
Vitamin D3
75 kIU/kg s.c.
Sampling
Day 1
Day 2
Day 3
Day 4
Day 5
Day 6
Day 7
Day 8
Day 9
Day 10
Day 11
Day 12
Treatment s.c.
G1: Saline
G2: 3 mg/kg SNF472
G3: 10 mg/kg SNF472
G4: 30 mg/kg SNF472
G5: 100 mg/kg SNF472

Figure 2. Experimental model for the analysis of SNF472 efficacy in the inhibition of cardiovascular calcification

- **Dose-response** in inhibition of calcification with **maximum inhibition** of **75 and 80%** in aorta and heart, respectively.

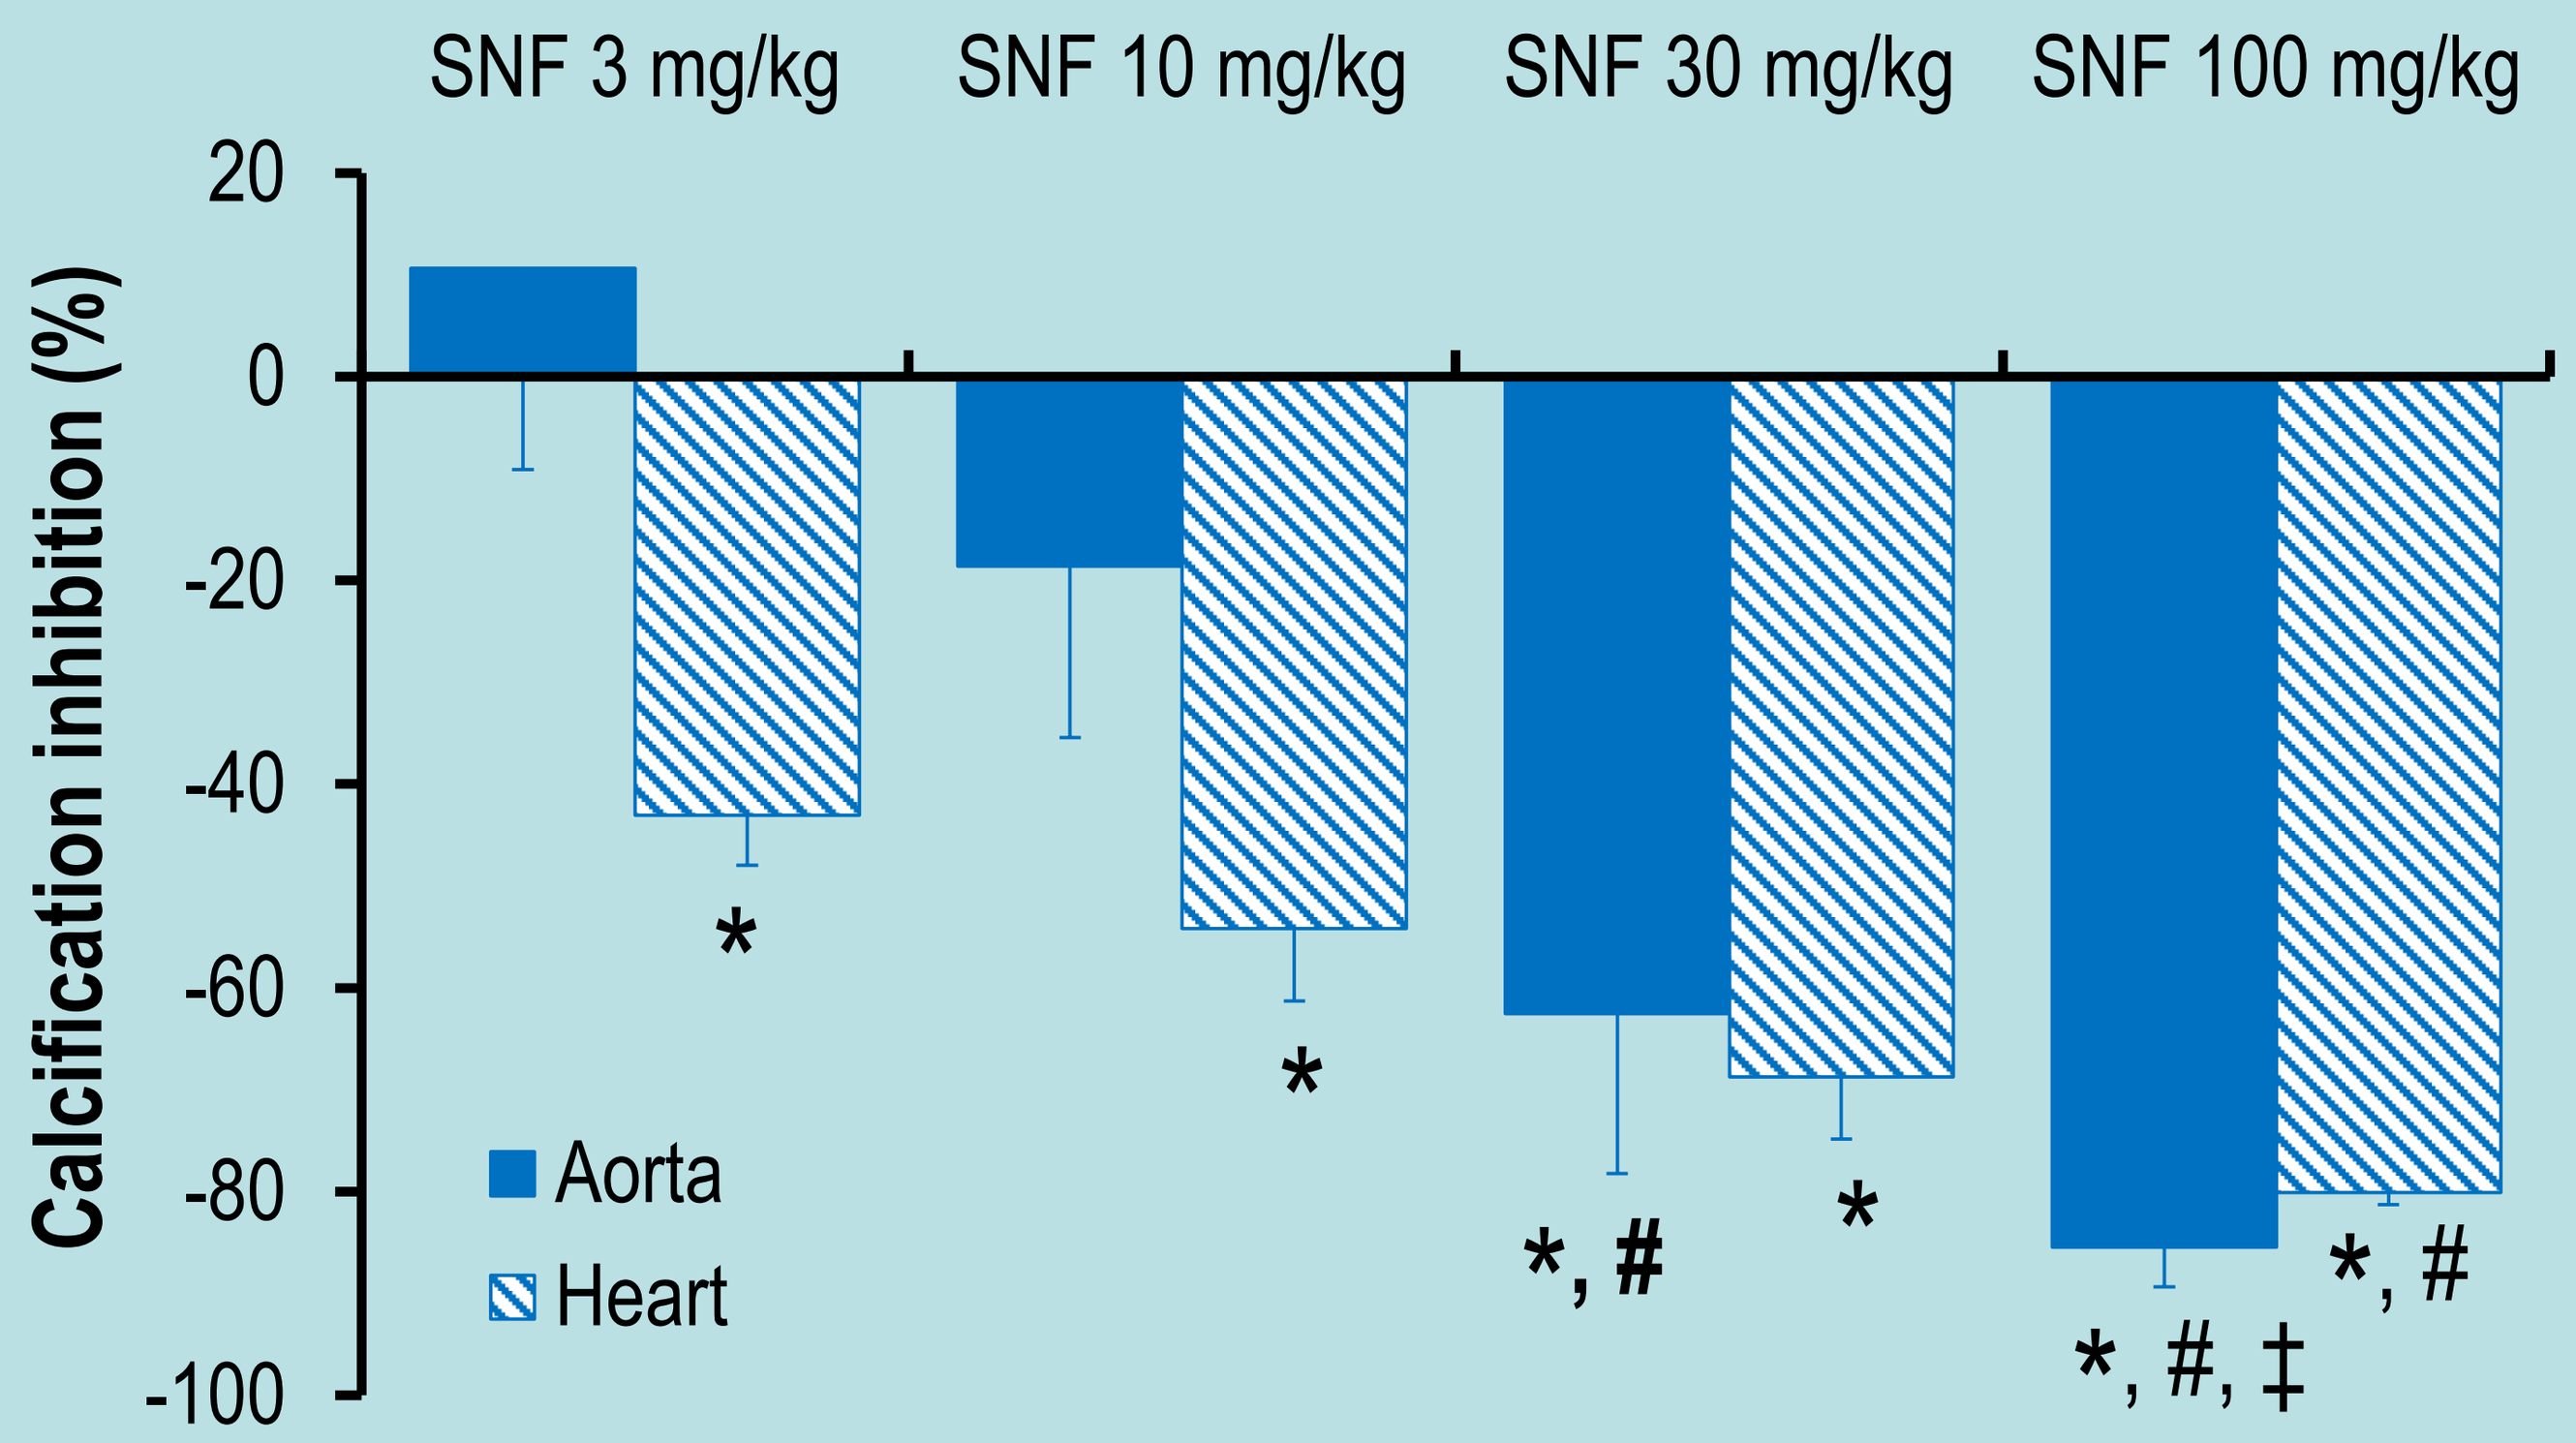
SNF 3 mg/kg
SNF 10 mg/kg
SNF 30 mg/kg
SNF 100 mg/kg
Calcification inhibition (%)
20
0
-20
-40
-60
-80
-100
Aorta
Heart
*
*
*, #
*
*, #, ‡
*, #

Figure 3. Inhibition of cardiovascular calcification by SNF472 in a rat model. Mean ± SEM. Statistical analysis: One-way ANOVA. (*) Differences vs control, (#) differences vs 3 mg/kg, (‡) differences vs 10 mg/kg; $p < 0.05$

FIRST-TIME-IN-HUMAN CLINICAL TRIAL

- Performed in eight hemodialysis patients.
- One single i.v. dose of 9 mg/kg SNF472 through dialysis tubing during the 4 hours of dialysis.
- **No systemic adverse events** nor effects in safety parameters.
- Pharmacokinetic measurements revealed a low SNF472 clearance through the dialysis membrane.

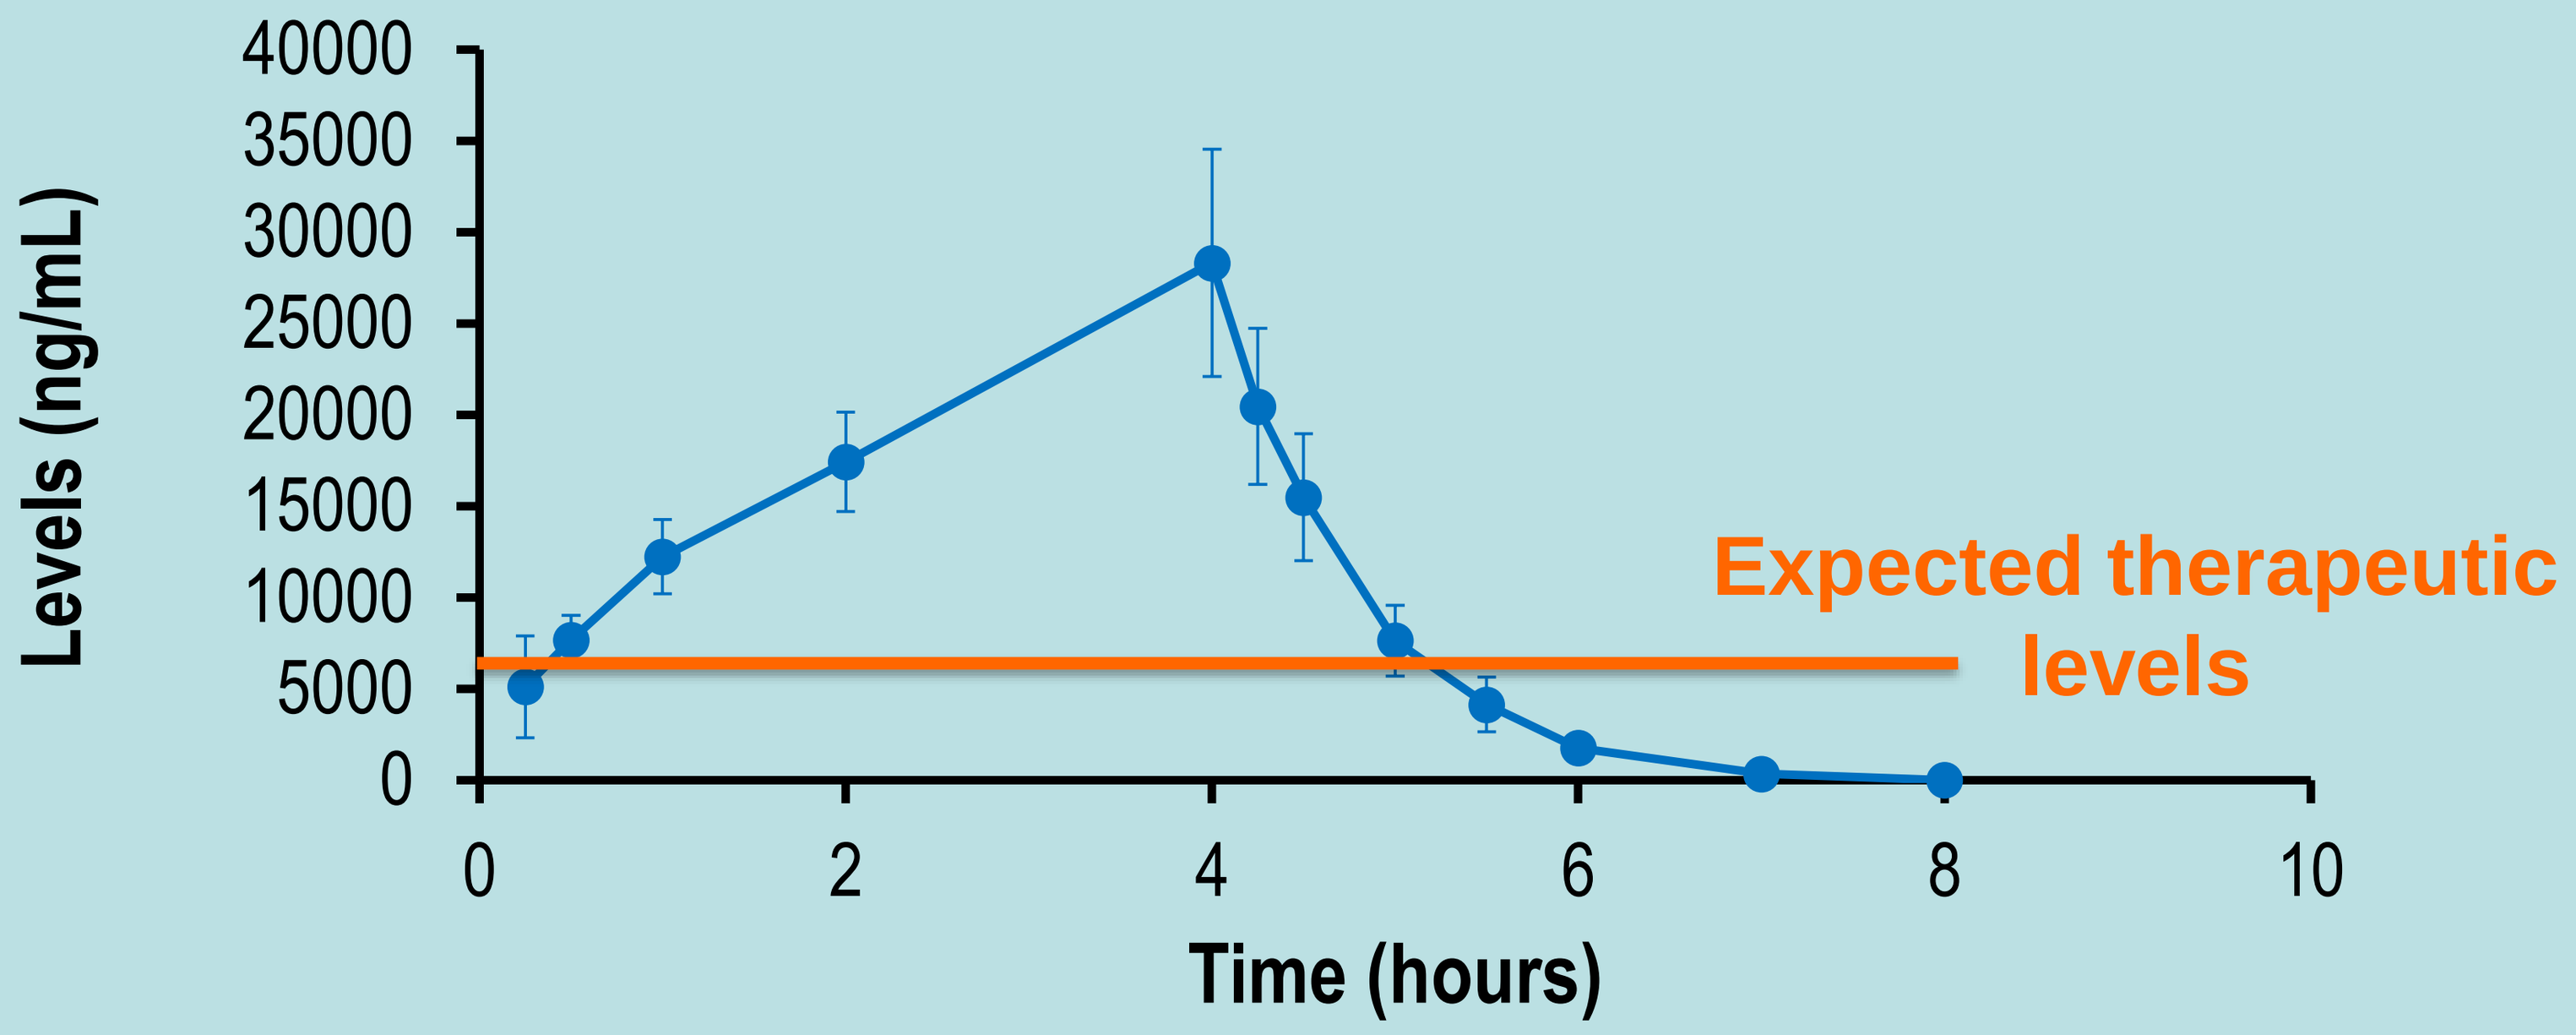
Levels (ng/mL)
40000
35000
30000
25000
20000
15000
10000
5000
0
0
2
4
6
8
10
Time (hours)
Expected therapeutic levels

Figure 4. Pharmacokinetics of 9 mg/kg SNF472 after 4 hour intravenous infusion in hemodialysis patients. Mean ± SEM.

- HAP crystallization was induced ex vivo using the method summarized in Figure 5A. **Induced HAP crystallization was inhibited up to 80%** in plasma samples obtained at the end of the SNF472 infusion in SNF472-treated HD patients.

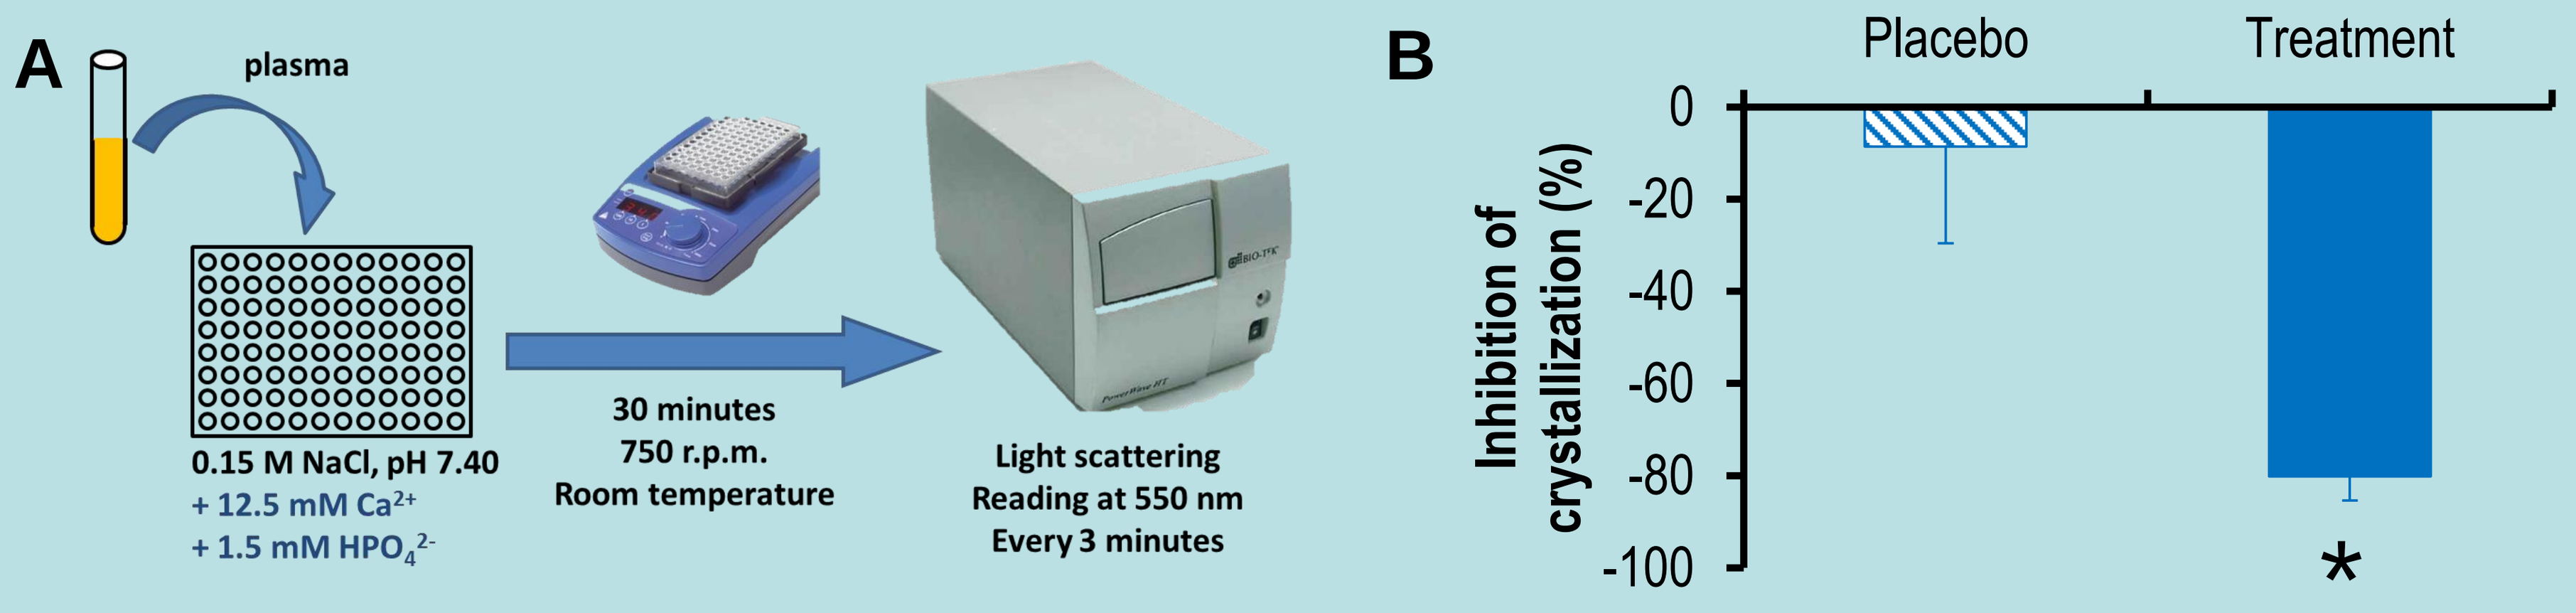
A
plasma
0.15 M NaCl, pH 7.40
+ 12.5 mM Ca2+
+ 1.5 mM HPO4 2-
30 minutes
750 r.p.m.
Room temperature
Light scattering
Reading at 550 nm
Every 3 minutes
B
Placebo
Treatment
Inhibition of crystallization (%)
0
-20
-40
-60
-80
-100
*

Figure 5. (A) Method to induce plasma hydroxyapatite crystallization ex vivo. (B) Ex-vivo inhibition of HAP crystallization by SNF472 in hemodialysis patients. Mean ± SEM.

CONCLUSIONS

1. **SNF472 inhibits CVC in animal models.**
2. **SNF472 inhibits HAP crystallization in human plasma, so it has the potential to inhibit calcification.**
3. **These results suggest a favorable benefit/risk ratio of SNF472 and support further studies in dialysis patients.**

This study was supported by RETOS COLABORACIÓN: RTC-2014-2460-1 ISCIII grant.